Identifying Adult Patients with Nonrelapsing Secondary Progressive Multiple Sclerosis Using Algorithms in US-based Healthcare Databases

Nupur Greene[1], Sarah N Gibbs[2], Michael S Broder[2], Lisa Farnett[1], Eunice Chang[2], Cynthia Campos[2], Keiko Higuchi[1]

[1]Sanofi, Cambridge, MA, USA; [2]PHAR, CA, USA

Ines Hemim (Ines.Hemim@sanofi.com) presenting on behalf of authors

BACKGROUND

- Multiple sclerosis (MS) is a chronic inflammatory, demyelinating, and degenerative disease of the central nervous system (CNS). It is categorized into phenotypes depending on whether the disease is relapsing (relapsing-remitting MS [RRMS]) or progressive (primary progressive MS [PPMS] or secondary progressive MS [SPMS]).[1,2]
- People with SPMS who still experience relapses are defined as having active SPMS (aSPMS).[2] In the US, disease modifying therapies (DMTs) approved for the treatment of RRMS can be used to treat aSPMS.
 - However, many patients with SPMS no longer experience relapses, which can be termed nonrelapsing SPMS (nrSPMS) and may not be benefiting from currently approved DMTs.
 - Given this unmet need and disease burden on these patients, an in-depth understanding of nrSPMS is important, particularly in the context of real-world evidence.
- While there is one ICD-10 code for MS, there are no codes for specific MS phenotypes, including none for SPMS overall or the nrSPMS subtype.

OBJECTIVE

This study aimed to develop a validated algorithm capable of identifying adult patients with nrSPMS in US-based electronic health records (EHR) or claims databases.

METHODS

- We developed algorithms capable of identifying patients with nrSPMS and tested them in two data sources – patient medical records (including billing records) and a large commercial database **(Figure 1)**

Figure 1. Flowchart of the process for algorithm development

Cognitive interviews with neurologists
- Three neurologist were interviewed to provide clinical input to develop potential candidate algorithms

Development of potential algorithms
- Based on clinical input from neurologists, candidate algorithms were developed

Study protocol & obtaining IRB approval
- Study protocol and data collection tool were developed to collect the data from patient medical records and clinic billing data
- The aim was to collect data on 200 adult patients with MS across 3 patient cohorts:
 - 100 patients with nrSPMS (as true positives)
 - 100 patients with either aSPMS or RRMS (as controls to serve as the comparison group)
- Central IRB approval was obtained

Study population, data collection from medical charts, and billing records
- Study population eligibility criteria included:
 - MS diagnosed patient ≥18 years
 - Patients last seen at the clinic no more than 2 years before the end of the study (i.e., IRB approval date 12/30/2021)
 - 3 years of available medical records, with at least 1 visit per calendar year
 - Physician diagnosis of nrSPMS (no clinical relapses in 2 years before index), aSPMS (≥1 relapse in the past 2 years before index) or RRMS at baseline
- De-identified patient data were collected from patient medical records and clinic billing records from various neurology sites
- All data were collected retrospectively (prior to the study end date of 12/30/2021)

Performance testing of algorithm candidates
- Tested the performance (sensitivity, specificity, positive predictive value [PPV], and negative predictive value [NPV]) of hundreds of versions of the algorithms in both medical records & clinical billing data of 195 patients

Face validity of the leading algorithms in US-based claims databases

Tested the face validity of the 2 best performing nrSPMS algorithms in a large US-based commercial claims database by observing whether the demographic, clinical, and utilization characteristics we would expect are found among patients identified with nrSPMS
- A retrospective cross-sectional analysis was conducted using IQVIA Pharmetrics Plus® database
- Descriptive statistics was conducted for the considered measures
- The measured characteristics were compared among patients with nrSPMS with known (i.e., expected) information derived from published clinical studies, other sources, and patient medical records collected in this study

aSPMS, active secondary progressive multiple sclerosis; DMT, disease modifying therapy; eCRF, electronic case report form; IRB, institutional review boards; NPV, negative predictive value; nrSPMS, nonrelapsing secondary progressive multiple sclerosis; PPV, positive predictive value; RRMS, relapsing-remitting multiple sclerosis.

RESULTS

- Candidate algorithms (8 clinically recommended algorithms and hundreds of variations based on exploratory analysis) made up of variables with existing ICD codes were developed to identify patients with nrSPMS in US-based healthcare claims datasets. Based on their performance in medical and billing records of 195 patients with MS across the US, 2 best-performing algorithms were identified to be further tested in IQVIA Pharmetrics Plus® claims database (2016-2020) **(Figure 2)**.
- In both medical/billing records, algorithm 1 resulted in 93%/92% sensitivity, 86%/90% PPV, 74%/84% specificity, and 87%/86% NPV, while algorithm 2 showed 93%/92% sensitivity, 76%/84% specificity, 86%/90% PPV, and 87%/86% NPV **(Table 1)**.

Figure 2. Most potential algorithms for assessing face validity in patient medical records

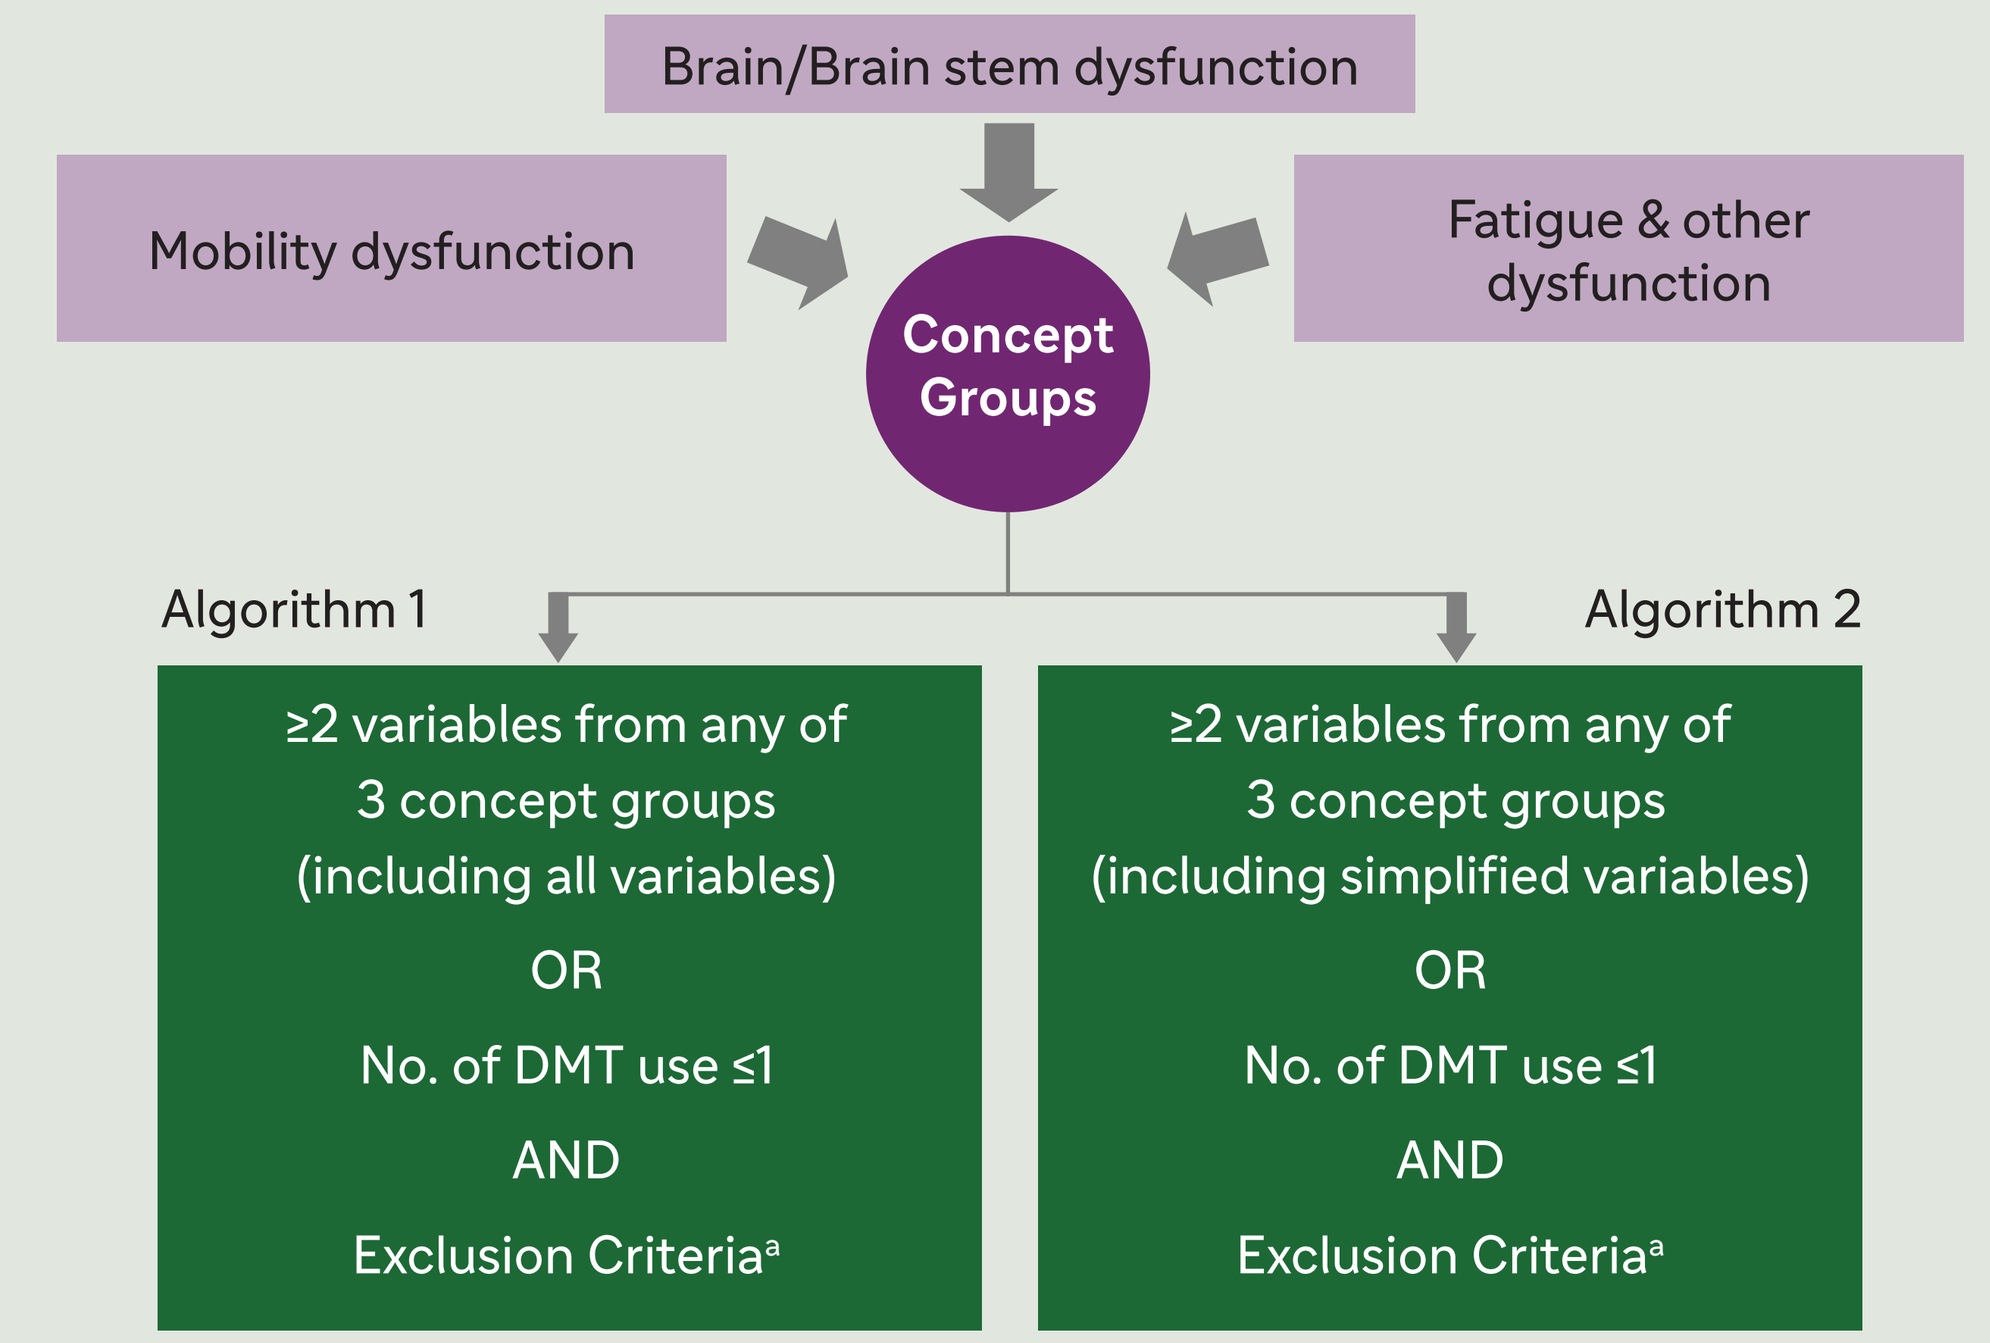
Brain/Brain stem dysfunction
Mobility dysfunction
Fatigue & other dysfunction
Concept Groups
Algorithm 1
Algorithm 2
≥2 variables from any of 3 concept groups (including all variables) OR No. of DMT use ≤1 AND Exclusion Criteria[a]
≥2 variables from any of 3 concept groups (including simplified variables) OR No. of DMT use ≤1 AND Exclusion Criteria[a]

Algorithm 1: ≥2 variables from any of 3 concept groups (mobility dysfunction, brain/brain stem dysfunction, other) OR (no. of DMT use ≤1 and no exclusions) Spinal cord dysfunction: Gait dysfunction AND use of dalfampridine (Ampyra); spasticity AND use of spasticity medication; use of ambulatory devices (e.g., cane, walker, wheelchair); physical therapy for ≥6 weeks in any 1-year period; occupational therapy for ≥6 weeks in any 1-year period; documented falls; ataxia, 2) Brain dysfunction: Neuropathic pain AND use of pain medication; trigeminal neuralgia AND use of pain medication; swallowing dysfunction (dysphagia); speech dysfunction (dysarthria); pseudobulbar affect AND use of Nuedexta (dextromethorphan/quinidine); optic neuritis; impaired cognition, and 3) Fatigue & Other dysfunction: Neurogenic bladder; neurogenic bowel; use of urinary catheter (e.g., self-catheterization, suprapubic catheter); bowel or bladder incontinence; in females - hospitalization for urinary tract infections (including acute cystitis, urosepsis, or kidney infection); hospitalization for respiratory infections; fatigue; insomnia; sleep apnea; sleep studies; circadian rhythm sleep disorder AND use of Provigil (modafinil) or Nuvigil (armodafinil).
Algorithm 2: Same as algorithm 1, but a shorter list of variables from the 3 concept groups. where, Mobility dysfunction: Use of spasticity medication, Use of ambulatory devices (e.g., cane, walker, wheelchair), Ataxia; Brain/brain stem dysfunction: Neuropathic pain AND use of pain medication, Trigeminal neuralgia, Speech dysfunction (dysarthria), Pseudobulbar affect, Optic neuritis, Impaired cognition; Fatigue & Other dysfunction: Neurogenic bladder, Use of urinary catheter (e.g., self-catheterization, suprapubic catheter), Bowel or bladder incontinence; Fatigue, Insomnia.
[a]Exclusion criteria : Age >70 years, OR Primary diagnosis of other neurological disorder (Alzheimer's, Parkinson's Disease, Myasthenia gravis, or stroke), OR ≥1 inpatient visit with a discharge diagnosis of multiple sclerosis (MS), OR ≥1 outpatient visit with a diagnosis of MS AND use of dexamethasone, methylprednisolone, prednisolone, prednisone, or adrenocorticotropin hormone on day of or within 7 days following the visit.
DMT, disease modifying therapy.

Table 1. Selected algorithm performance in EHR

Data source	Algorithm	Sensitivity	PPV	Specificity	NPV
Medical records	Algorithm 1	93%	86%	74%	87%
	Algorithm 2	93%	86%	76%	87%
Billing records (assuming all inpatient visits and medications matched the data in the medical records)	Algorithm 1	92%	90%	84%	86%
	Algorithm 2	92%	90%	84%	86%

EHR, electronic health record; NPV, negative predictive value; PPV, positive predictive value.

- A total of 33,244 MS patients were identified in the IQVIA database between 1/1/2016 and 12/31/2018. A random MS claim during this period was selected as the start date, and 2 years observation period since the start date were used for algorithm identification.
 - After applying additional algorithm-specific criteria, the total nrSPMS patients identified by algorithm 1 were 19,661 patients and algorithm 2 were 19,783 patients **(Figure 3)**.

Figure 3. Algorithms attrition chart in IQVIA database

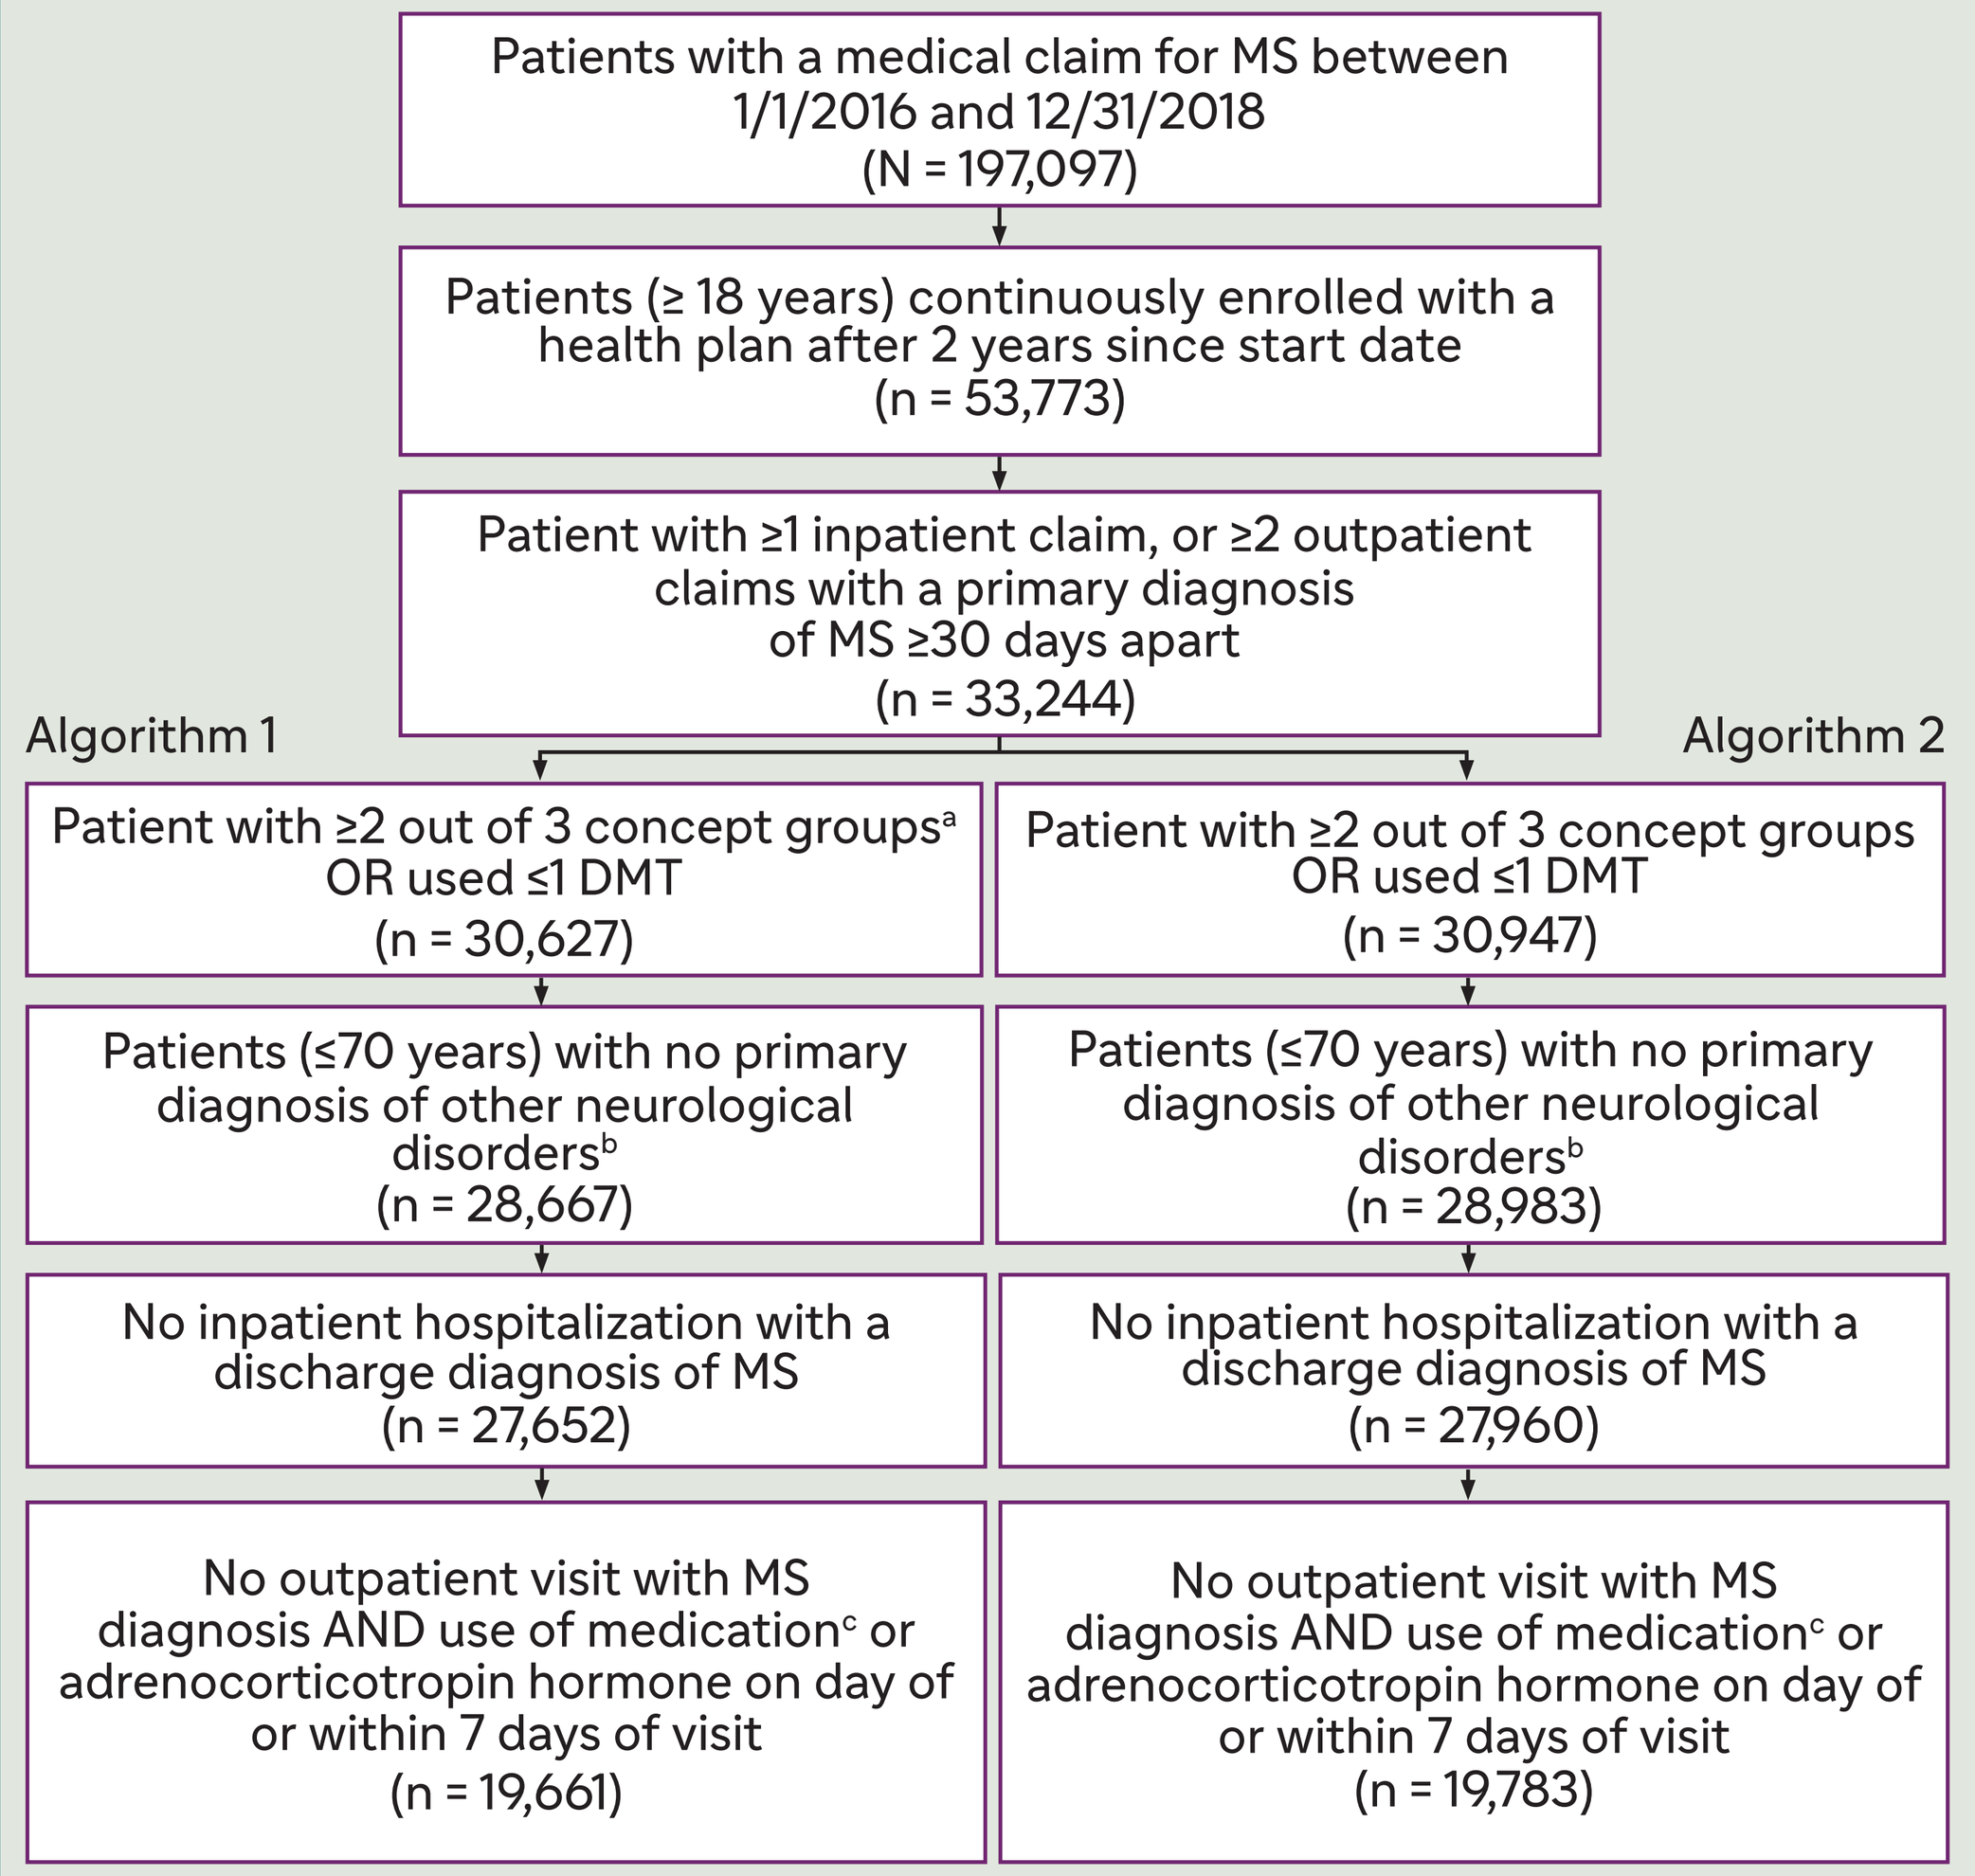
Patients with a medical claim for MS between 1/1/2016 and 12/31/2018 (N = 197,097)
Patients (≥ 18 years) continuously enrolled with a health plan after 2 years since start date (n = 53,773)
Patient with ≥1 inpatient claim, or ≥2 outpatient claims with a primary diagnosis of MS ≥30 days apart (n = 33,244)
Algorithm 1
Algorithm 2
Patient with ≥2 out of 3 concept groups[a] OR used ≤1 DMT (n = 30,627)
Patient with ≥2 out of 3 concept groups OR used ≤1 DMT (n = 30,947)
Patients (≤70 years) with no primary diagnosis of other neurological disorders[b] (n = 28,667)
Patients (≤70 years) with no primary diagnosis of other neurological disorders[b] (n = 28,983)
No inpatient hospitalization with a discharge diagnosis of MS (n = 27,652)
No inpatient hospitalization with a discharge diagnosis of MS (n = 27,960)
No outpatient visit with MS diagnosis AND use of medication[c] or adrenocorticotropin hormone on day of or within 7 days of visit (n = 19,661)
No outpatient visit with MS diagnosis AND use of medication[c] or adrenocorticotropin hormone on day of or within 7 days of visit (n = 19,783)

[a]concept groups: Spinal cord dysfunction, brain dysfunction, other (neurogenic bladder, neurogenic bowel, fatigue, insomnia, etc);
[b]Alzheimer's, Parkinson's Disease, Myasthenia gravis, or stroke;[c]dexamethasone, methylprednisolone, prednisolone, prednisone.
N, number of patients; DMT, disease modifying therapy; MS, multiple sclerosis.

- Demographic, clinical, and utilization characteristics of these patients were reported in **Table 2.**

Table 2. Demographics and clinical characteristics of patients with nrSPMS

	Algorithm 1 (N = 19,661)	Algorithm 2 (N = 19,783)
Age, year, Mean (SD)	48.6 (10.5)	48.5 (10.5)
Female	14,903 (75.8)	14,998 (75.8)
Geographic region		
Midwest	5,814 (29.6)	5,840 (29.5)
Northeast	4,381 (22.3)	4,419 (22.3)
South	6,484 (33.0)	6,527 (33.0)
West	2,982 (15.2)	2,997 (15.1)
Insurance status		
Commercial	11,958 (60.8)	12,024 (60.8)
Medicaid	198 (1.0)	198 (1.0)
Medicare	1,326 (6.7)	1,334 (6.7)
Other/Unknown	6,179 (31.4)	6,227 (31.5)

Table 2. (Cont'd)

	Algorithm 1 (N = 19,661)	Algorithm 2 (N = 19,783)
Clinical Characteristics		
Gait dysfunction	3,527 (17.9)	3,534 (17.9)
Fatigue	6,437 (32.7)	6,505 (32.9)
Spasticity	2,240 (11.4)	2,228 (11.3)
Bowel or bladder incontinence	3,265 (16.6)	3,304 (16.7)
Optic neuritis	1,815 (9.2)	1,825 (9.2)
Insomnia	1,715 (8.7)	1,742 (8.8)
Use of ambulatory devices (e.g., cane, walker, wheelchair)	1,087 (5.5)	1,083 (5.5)
DMT	14,433 (73.4)	14,555 (73.6)
Glatiramer acetate	3,529 (17.9)	3,573 (18.1)
Dimethyl fumarate	3,074 (15.6)	3,117 (15.8)
Beta interferon	2,613 (13.3)	2,645 (13.4)
Fingolimod	1,873 (9.5)	1,898 (9.6)
Other Medications Use		
Pain medication	4,947 (25.2)	5,004 (25.3)
Spasticity medication	5,930 (30.2)	6,101 (30.8)
Comorbidity Conditions of Interest		
Multiple sclerosis comorbidity	15,370 (78.2)	15,489 (78.3)
Burning/numbness/tingling	3,766 (19.2)	3,781 (19.1)
Healthcare utilization		
LOS (days) per patient among utilizers	1,853 (7.57)	1,864 (7.52)
Inpatient hospitalizations	1,853 (9.4)	1,864 (9.4)
Receiving ICU care	278 (1.4)	280 (1.4)
Any ED visits	3,877(19.7)	3,904 (19.7)
No. of outpatient hospital visits, mean (SD)	7.5 (10.8)	7.5 (10.8)

All data are presented as n (%) unless otherwise mentioned.
N, number of patients; DMT, disease modifying therapy; ED, emergency department; ICU, intensive care unit; LOS, length of stay; nrSPMS, nonrelapsing secondary progressive multiple sclerosis; SD, standard deviation

- To assess the face validity of the above 2 algorithms, characteristics of the patients identified using these algorithms were compared to published clinical studies and with patient medical records collected in this study. The characteristics were consistent, indicating that both algorithms 1 & 2 had face validity.
- While both algorithms 1 and 2 were specific (patients identified with either are likely to have nrSPMS), algorithm 2 missed fewer patients with nrSPMS in IQVIA database compared to algorithm 1.

LIMITATIONS

- Algorithm performance may be influenced by the quality of the data source used. For example in some data sources, characteristics may be recorded inconsistently or incompletely, causing some potential misclassification of patients.
- While some items included may increase the sensitivity of the algorithm, they may also decrease the specificity.
- Cost was not included in the algorithms as it was not identified as a significant criteria in identifying nrSPMS patients and future studies using our algorithms will likely explore healthcare cost.

CONCLUSIONS

- The proposed algorithms showed high performance when tested in patient medical record data.
- Additionally, the algorithms identified a cohort of patients in claims data that appeared consistent with clinically identified patients with nrSPMS (based on inclusion/exclusion criteria).
- These algorithms can be applied in other US EHR or claims-based datasets to facilitate further research to better identify and describe the nrSPMS population.

This QR code directs to previous presentation at ISPOR annual meeting 2023, and is for personal use only

Disclosures

IH (Presenter), NG, LF, and KH are employees of Sanofi and may hold stock/stock-options in Sanofi.
SNG, MSB, EC, and CC are employees of PHAR, which was paid by Sanofi to conduct the research described in this poster. PHAR also discloses financial relationships with the following entities outside of the submitted work: Akcea, Amgen, Boston Scientific Corporation, Eisai, Genentech, Greenwich Biosciences, Ionis, Jazz, Novartis, Prothena, Recordati, Regeneron, Sanofi, Sunovion, and Takeda Pharmaceuticals.

Acknowledgments

Medical writing support for original poster and editorial assistance for this encore poster were provided by Chiranjit Ghosh, PhD and Sanjeev Kallapari of Sanofi.
Data included in this poster were originally presented at the ISPOR Annual Meeting, Boston, MA, USA; May 07–10, 2023.

Funding

This study was funded by Sanofi.

References

1. Lublin FD, et al. *Neurology.* 2014;83:278-86.
2. Klineova S, Lublin FD. *Cold Spring Harb Perspect Med.* 2018;8.

Poster presented at Academy of Managed Care Pharmacy (AMCP) Nexus, Orlando, FL, USA; October 16–19, 2023.