PHARMACOLOGICAL TREATMENT PATTERNS AFTER INITIATION OF ORAL CARBIDOPA/LEVODOPA AMONG PATIENTS WITH PARKINSON'S DISEASE

Andrew Thach,[1] Sheila Reiss Reddy,[2] Eunice Chang,[2] Marian H. Tarbox,[2] Darshan Mehta,[1] Alyssa Bowling,[1] Jill Farmer[3]

[1]Sunovion Pharmaceuticals Inc., Marlborough, Massachusetts, United States; [2]Partnership for Health Analytic Research (PHAR), LLC, Beverly Hills, California, United States; [3]Parkinson's Disease & Movement Disorder Program, Global Neurosciences Institute and Drexel University College of Medicine, Philadelphia, Pennsylvania, United States

KEY FINDINGS

- In this retrospective cohort study of claims data from a large United States insurance database, many patients with Parkinson's disease (PD) who initiated treatment with oral carbidopa/levodopa (CD/LD) required adjustments to baseline CD/LD and/or concomitant medications over ≥1 year of follow-up
- Approximately two-thirds of all patients had an increase in treatment intensity during follow-up, defined as an increase in the total daily levodopa dose or new concomitant PD medication use
- One-third of patients used concomitant PD medications throughout follow-up, with dopamine agonists being the most common; one-quarter of those patients switched to or added a second class of concomitant medication, with dopamine agonist add-on combinations being the most frequent
- Various treatment approaches are available to maintain symptom control in patients with PD who have initiated CD/LD. Clinicians should closely monitor patients as the disease progresses and adjust regimens as needed, considering all available options

Supported by funding from Sunovion Pharmaceuticals Inc.

Poster presented at the International Society for Pharmacoeconomics and Outcomes Research (ISPOR); May 15–18, 2022; Washington, DC, United States

INTRODUCTION

- PD is a chronic, progressive neurologic disorder resulting from degeneration of dopamine neurons in the substantia nigra that manifests as motor (eg, rest tremor, rigidity, bradykinesia, and postural instability) and nonmotor (eg, olfactory loss, sleep dysfunction, autonomic dysfunction, psychiatric disturbances, and cognitive impairment) symptoms[1,2]
- Oral CD/LD is often the mainstay treatment of motor symptoms for PD[2,3]
- Motor symptoms initially improve with lower doses of CD/LD[4]; however, patients usually require adjustments to their baseline regimen to maintain symptom control over time because of disease progression[2]
- Treatment approaches to improve motor symptom control include the following:
 - Increasing CD/LD daily dose or dosing frequency[2]
 - Adding dopamine agonists, monoamine oxidase-B inhibitors, catechol-O-methyltransferase inhibitors, istradefylline, or amantadine[1,5]
 - Adding acute, intermittent treatments, such as apomorphine subcutaneous injection, apomorphine sublingual film, or levodopa inhalation powder[3]
 - Initiating advanced therapies (eg, continuous levodopa infusion, deep brain stimulation, etc.)[2]

OBJECTIVE

- Describe treatment patterns in patients with PD who are newly treated with CD/LD

METHODS

Study Design and Analyses

- This retrospective cohort study of patients with PD used de-identified data from a large United States insurance database
- Patients with PD (≥1 inpatient or ≥2 outpatient claims with International Classification of Diseases, Ninth Revision, Clinical Modification [332.0] or Tenth Revision, Clinical Modification [G20.xx] codes) were identified
- Among these patients, the following inclusion criteria also applied:
 - The index date was the date of the first observed claim for oral CD/LD medication from 01 Jan 2016 to 31 Dec 2019
 - Patients were lacking CD/LD claims during the 1-year pre-index period (baseline) but may have been taking dopamine agonists
 - Patients were continuously enrolled during baseline and ≥1 year after the index date (follow-up)
- Treatment patterns were evaluated during baseline and during the ≥1-year follow-up period for the main cohort of patients (N=16,076) and a subset who used concomitant PD medications during follow-up (n=5092)
- Treatment patterns that were assessed included increase in treatment intensity (increase in total daily levodopa dose or new concomitant PD medication use), any use of acute, intermittent treatments, and switch/add-on patterns in patients using concomitant PD medications

Statistical Analyses

- Descriptive statistics were used to evaluate patient characteristics and treatment use
- All data transformations and statistical analyses were performed using SAS® version 9.4

RESULTS

Patients

- A total of 16,076 patients with PD who were newly treated with CD/LD were identified (**Figure 1**)

Figure 1. Cohort Selection

Patients who had ≥1 inpatient or ≥2 outpatient claims of PD diagnosis during the study period (2015–2020)
(N=156,423)

↓

Patients who filled an oral CD/LD medication in the identification period (2016–2019)[a]
(n=79,200)

↓

Patients who were continuously enrolled in the 1-year pre-index and 1-year post-index periods
(n=38,599)

↓

Patients who had ≥1 PD diagnosis before or on the index date
(n=32,718)

↓

Patients who were treatment-naïve (no use of CD/LD during the baseline period)
(n=17,079)

↓

Patients who had no ambiguous multiple oral CD/LD claims filled on the same day on the index date or during the follow-up period
(n=16,076)

[a]The first fill date in the identification period was defined as the index date.
CD/LD, carbidopa/levodopa; PD, Parkinson's disease.

- Mean (standard deviation [SD]) age was 75.3 (8.3) years, 40% of patients were female, and 66% were White
- Mean (SD) number of chronic conditions was 6.0 (2.4), and 46% of patients had evidence of frailty,[6] with falls (24%), frailty with advanced illness or dementia (23%), and fractures (11%) being the most common (**Table 1**)

Table 1. Baseline Frailty Indicators

Characteristic	Patients Newly Treated With CD/LD (N=16,076)
Frailty indicators, %	46
Falls	24
Frailty (with advanced illness or dementia)	23
Fracture	11
Home oxygen use	6
Walking aid use	6
Wheelchair use	4
Urinary catheter use	3
Weighted vest	<1

CD/LD, carbidopa/levodopa.

- Mean (SD) time from first observed PD diagnosis to the index treatment was 125.9 (153.7) days
- Insurance coverage among patients was predominantly Medicare (89%); 11% of patients had commercial health insurance

Index Treatment and Treatment Patterns During the Baseline Period

- The most commonly used index CD/LD formulation was the 25/100-mg oral tablet in 82% of patients
- A total of 17% of patients used other PD medications during the baseline period before initiating CD/LD, with dopamine agonists being the most common (**Table 2**)

Table 2. Use of Other PD Medications During the Baseline and Follow-up Periods

Medication	Patients Newly Treated With CD/LD (N=16,076)	
	Treatment With Other PD Medication in the Baseline Period Before Initiating CD/LD	Concomitant Therapy During the Follow-up Period
Patients using other PD medications, %	17	32
Dopamine agonists	11	20
MAO-B inhibitors	6	10
Amantadine	3	8
COMT inhibitors	<1	3

CD/LD, carbidopa/levodopa; COMT, catechol-O-methyltransferase; MAO-B, monoamine oxidase-B; PD, Parkinson's disease.

Treatment Patterns During the Follow-up Period

- The median duration of the follow-up period was 846 days
- Median duration of index CD/LD treatment (ie, time from index date until gap in treatment of ≥30 days) was 238 days
- A total of 32% of patients used other PD medications concomitant to CD/LD during the follow-up period, with dopamine agonists being the most common (**Table 2**)
- An increase in treatment intensity was observed in 65% of patients (**Table 3**)

Table 3. Treatment Patterns During the Follow-up Period

Characteristic	Patients Newly Treated With CD/LD (N=16,076)
Patients with increase in treatment intensity (increase in total daily levodopa dose or new concomitant PD medication use), %	65
Days to increase in treatment intensity among patients with event, median	111
Increase in total daily levodopa dose, %	58
Total daily levodopa dose increase, mg, median	150
Days to total daily levodopa dose increase among patients with event, median	127
New concomitant PD medication use, %	23
Days to new concomitant PD medication use among patients with event, median	212
Any use of acute, intermittent treatments, %	<1
Apomorphine subcutaneous injection, %	<1
Apomorphine sublingual film, %	<1
Levodopa inhalation powder, %	<1
Days to acute, intermittent treatment use among patients with event, median	637

CD/LD, carbidopa/levodopa; PD, Parkinson's disease.

- Less than 1% of patients initiated acute, intermittent treatments
- Approximately 1% of patients initiated CD/LD administration via percutaneous endoscopic gastrostomy with jejunal extension, 1% initiated deep brain stimulation, and 3% initiated botulinum toxin therapy
- Among 5092 patients who used concomitant PD medications during follow-up, 24% switched to/added a second class of concomitant medication (median of 133 days until event); dopamine agonist add-on combinations (with monoamine oxidase-B inhibitors or amantadine) were most frequently used during follow-up (**Table 4**)

Table 4. Switch/Add-on[a] Treatment Patterns in Patients Who Used Concomitant PD Medications During the Follow-up Period

Characteristic	Patients Newly Treated With CD/LD Who Used Concomitant PD Medication (N=5092)
Switch/add-on concomitant PD medication, %	24
Days to the second (switch/add-on) concomitant PD medication among patients with event, median	133
Switch concomitant PD medication, %	8
Days to switch to the second concomitant PD medication among patients with event, median	220
Add-on concomitant PD medication, %	16
Days to add-on of the second concomitant PD medication among patients with event, median	95
Most Common (≥5%) First and Second Concomitant PD Medication Treatment Patterns,[b] %	**n=1227**
Dopamine agonists + MAO-B inhibitors	18
MAO-B inhibitors + dopamine agonists	13
Dopamine agonists + amantadine	12
Amantadine + dopamine agonists	8
Dopamine agonists → amantadine	6
MAO-B inhibitors + amantadine	6
Amantadine → dopamine agonists	5
MAO-B inhibitors → dopamine agonists	5

[a]After starting a new class of concomitant medication, if there is no refill of the previous concomitant medication, this event is defined as switch; otherwise (with refills of previous medication), the event is defined as add-on.
[b]Switch denoted by "→"; add-on denoted by "+".
CD/LD, carbidopa/levodopa; MAO-B, monoamine oxidase-B; PD, Parkinson's disease.

DISCLOSURES AND ACKNOWLEDGMENTS

SRR, EC, MHT, are employees of PHAR, LLC, a health services research company hired by Sunovion Pharmaceuticals Inc. (Marlborough, Massachusetts, United States) to conduct the research presented in this poster. JF is a consultant for Sunovion Pharmaceuticals Inc. AT, DM, and AB are employees of Sunovion Pharmaceuticals Inc. Medical writing and editorial assistance were provided by Edwin Thrower, PhD, CMPP, and Robert Schupp, PharmD, CMPP, of The Lockwood Group (Stamford, CT) and were supported by funding from Sunovion Pharmaceuticals Inc.

REFERENCES

1. Olanow CW, et al. *Neurology*. 2009;72(21 suppl 4):S1-S136. **2.** Armstrong MJ and Okun MS. *JAMA*. 2020;323:548-560. **3.** Olanow CW, et al. *Mov Disord*. 2021;36:2244-2253. **4.** Chou KL, et al. *Parkinsonism Relat Disord*. 2018;51:9-16. **5.** Shang P, et al. *J Mov Disord*. 2021;14:193-203. **6.** Izurieta HS, et al. *J Infect Dis*. 2019;220:1255-64.