Healthcare Utilization and Costs in Commercially Insured Patients With AL Amyloidosis

Tiffany P. Quock,[1] Jessie Tingjian Yan,[2] Eunice Chang,[2] Spencer D. Guthrie,[3] Michael S. Broder[2]

[1]Prothena Biosciences Inc, South San Francisco, CA, USA; [2]Partnership for Health Analytic Research, LLC, Beverly Hills, CA, USA; [3]San Francisco, CA, USA

4659

INTRODUCTION

- Amyloid light chain (AL) amyloidosis is a rare, progressive, and typically fatal disease caused by extracellular deposition of misfolded immunoglobulin light chains[1-3]
- Soluble toxic aggregates and deposited fibrils (amyloid) lead to progressive failure of vital organs, including the heart, kidneys, and nervous system, causing significant morbidity and mortality[1-3]
- The economic burden of AL amyloidosis has not been well characterized[4]

OBJECTIVE

- To examine healthcare utilization and costs associated with AL amyloidosis in the United States using real-world, nationally representative health insurance claims data

METHODS

Study Design and Data Source

- Retrospective, cross-sectional study using Truven MarketScan® Commercial and Medicare Supplement Insurance databases
 - These databases cover approximately 65 million commercially insured persons and their dependents and 5.3 million Medicare-eligible retired persons
- An algorithm was developed to identify patients with AL amyloidosis given the lack of specific diagnostic codes for this disease (detailed in Study Population below)

Study Population

- Patients were identified separately by each calendar year
- Patients aged ≥18 years with AL amyloidosis were identified if they
 - Filed ≥1 inpatient claim or ≥2 outpatient claims consistent with AL amyloidosis (*International Classification of Diseases, Ninth Revision, Clinical Modification* [*ICD-9-CM*] code 277.30 or 277.39; *International Classification of Diseases, Tenth Revision, Clinical Modification* [*ICD-10-CM*] code E85.4x, E85.8x, or E85.9x) in any diagnosis field

 AND
 - Received 1 AL amyloidosis–specific treatment (eg, chemotherapy, hematopoietic stem cell transplant [HSCT]) on or after the first amyloidosis diagnosis
- Patients enrolled with a healthcare plan from the beginning of each calendar year to the end of enrollment or the end of the calendar year, whichever occurred first

Study Measures

- All-cause healthcare utilization in each calendar year, including inpatient hospitalization, emergency department (ED) visit, non–ED outpatient service visit, and any treatment
- Total all-cause healthcare costs were calculated by adding inpatient, ED outpatient, non–ED outpatient, and pharmacy costs

Statistical Analysis

- Mean ± standard deviation (SD) were reported for continuous data
- Relative frequencies and percentages were reported for categorical data
- Mean healthcare utilization and costs were reported per patient without annualization
- Cost estimates were converted to 2015 US dollars using the Consumer Price Index to adjust for inflation
- Data transformations and statistical analyses were performed using SAS® version 9.4

RESULTS

Patient Demographics

- The overall study sample included 7326 patients (368-1080 unique patients per year, from 2007 to 2015)
 - Mean (±SD) age was 63.6 (±12.1), 45% of patients were female, all US regions were represented, and most patients had commercial insurance and preferred provider organization plans (**Table 1**)

Table 1. Patient Demographic Characteristics and Insurance Types by Calendar Year

Demographics	2007	2008	2009	2010	2011	2012	2013	2014	2015	All
N	349	573	713	792	972	1059	1031	1031	806	7326
Age, years, mean (SD)	63.6 (11.9)	63.1 (11.4)	62.2 (11.8)	62.4 (12.4)	63.9 (12.4)	63.8 (12.1)	65.0 (12.2)	63.6 (11.8)	64.1 (12.7)	63.6 (12.1)
Age category, years, %										
18-34	1.1	0.7	0.7	2.1	1.5	1.3	0.7	1.1	1.5	1.2
35-64	54.2	59.7	61.6	58.1	53.8	53.8	50.6	61.6	58.1	53.8
≥65	44.7	39.6	37.7	39.8	44.7	44.9	48.7	42.6	43.9	43.3
Female, %	45.8	46.2	46.8	45.3	45.4	42.7	43.7	44.3	47.3	45.0
Region, %										
Midwest	34.7	31.2	33.0	28.3	28.0	23.6	23.1	23.4	23.6	26.6
Northeast	12.6	15.7	18.1	20.5	22.3	25.0	22.9	26.7	24.9	22.1
South	37.8	35.6	30.4	33.0	30.2	32.7	33.1	36.3	36.6	33.6
West	14.9	17.5	18.5	18.3	19.4	18.7	21.0	13.7	14.9	17.6
Insurance type, %										
Commercial	56.2	62.1	64.4	61.7	56.6	56.2	52.3	58.6	57.8	58.1
Medicare Supplement Insurance[a]	43.8	37.9	35.6	38.3	43.4	43.8	47.7	41.4	42.2	41.9
Plan type, %										
PPO	51.0	53.4	51.8	49.0	55.1	55.0	52.8	53.1	54.5	53.1
Other	49.0	46.4	48.3	51.1	44.8	45.1	47.3	46.9	45.7	46.9

PPO, preferred provider organization.

[a]Medicare Supplement Insurance (Medigap) policy, sold by private companies, can help pay some of the healthcare costs that Original Medicare does not cover, such as copayments, coinsurance, and deductibles.

Treatment Utilization

- The use of bortezomib increased during the study period, from 4.6% in 2007 to 25.3% in 2015; in contrast, the use of melphalan decreased during the study period, from 18.9% in 2007 to 2.0% in 2015 (**Figure 1**)

Figure 1. Treatment utilization[a,b] by calendar year.

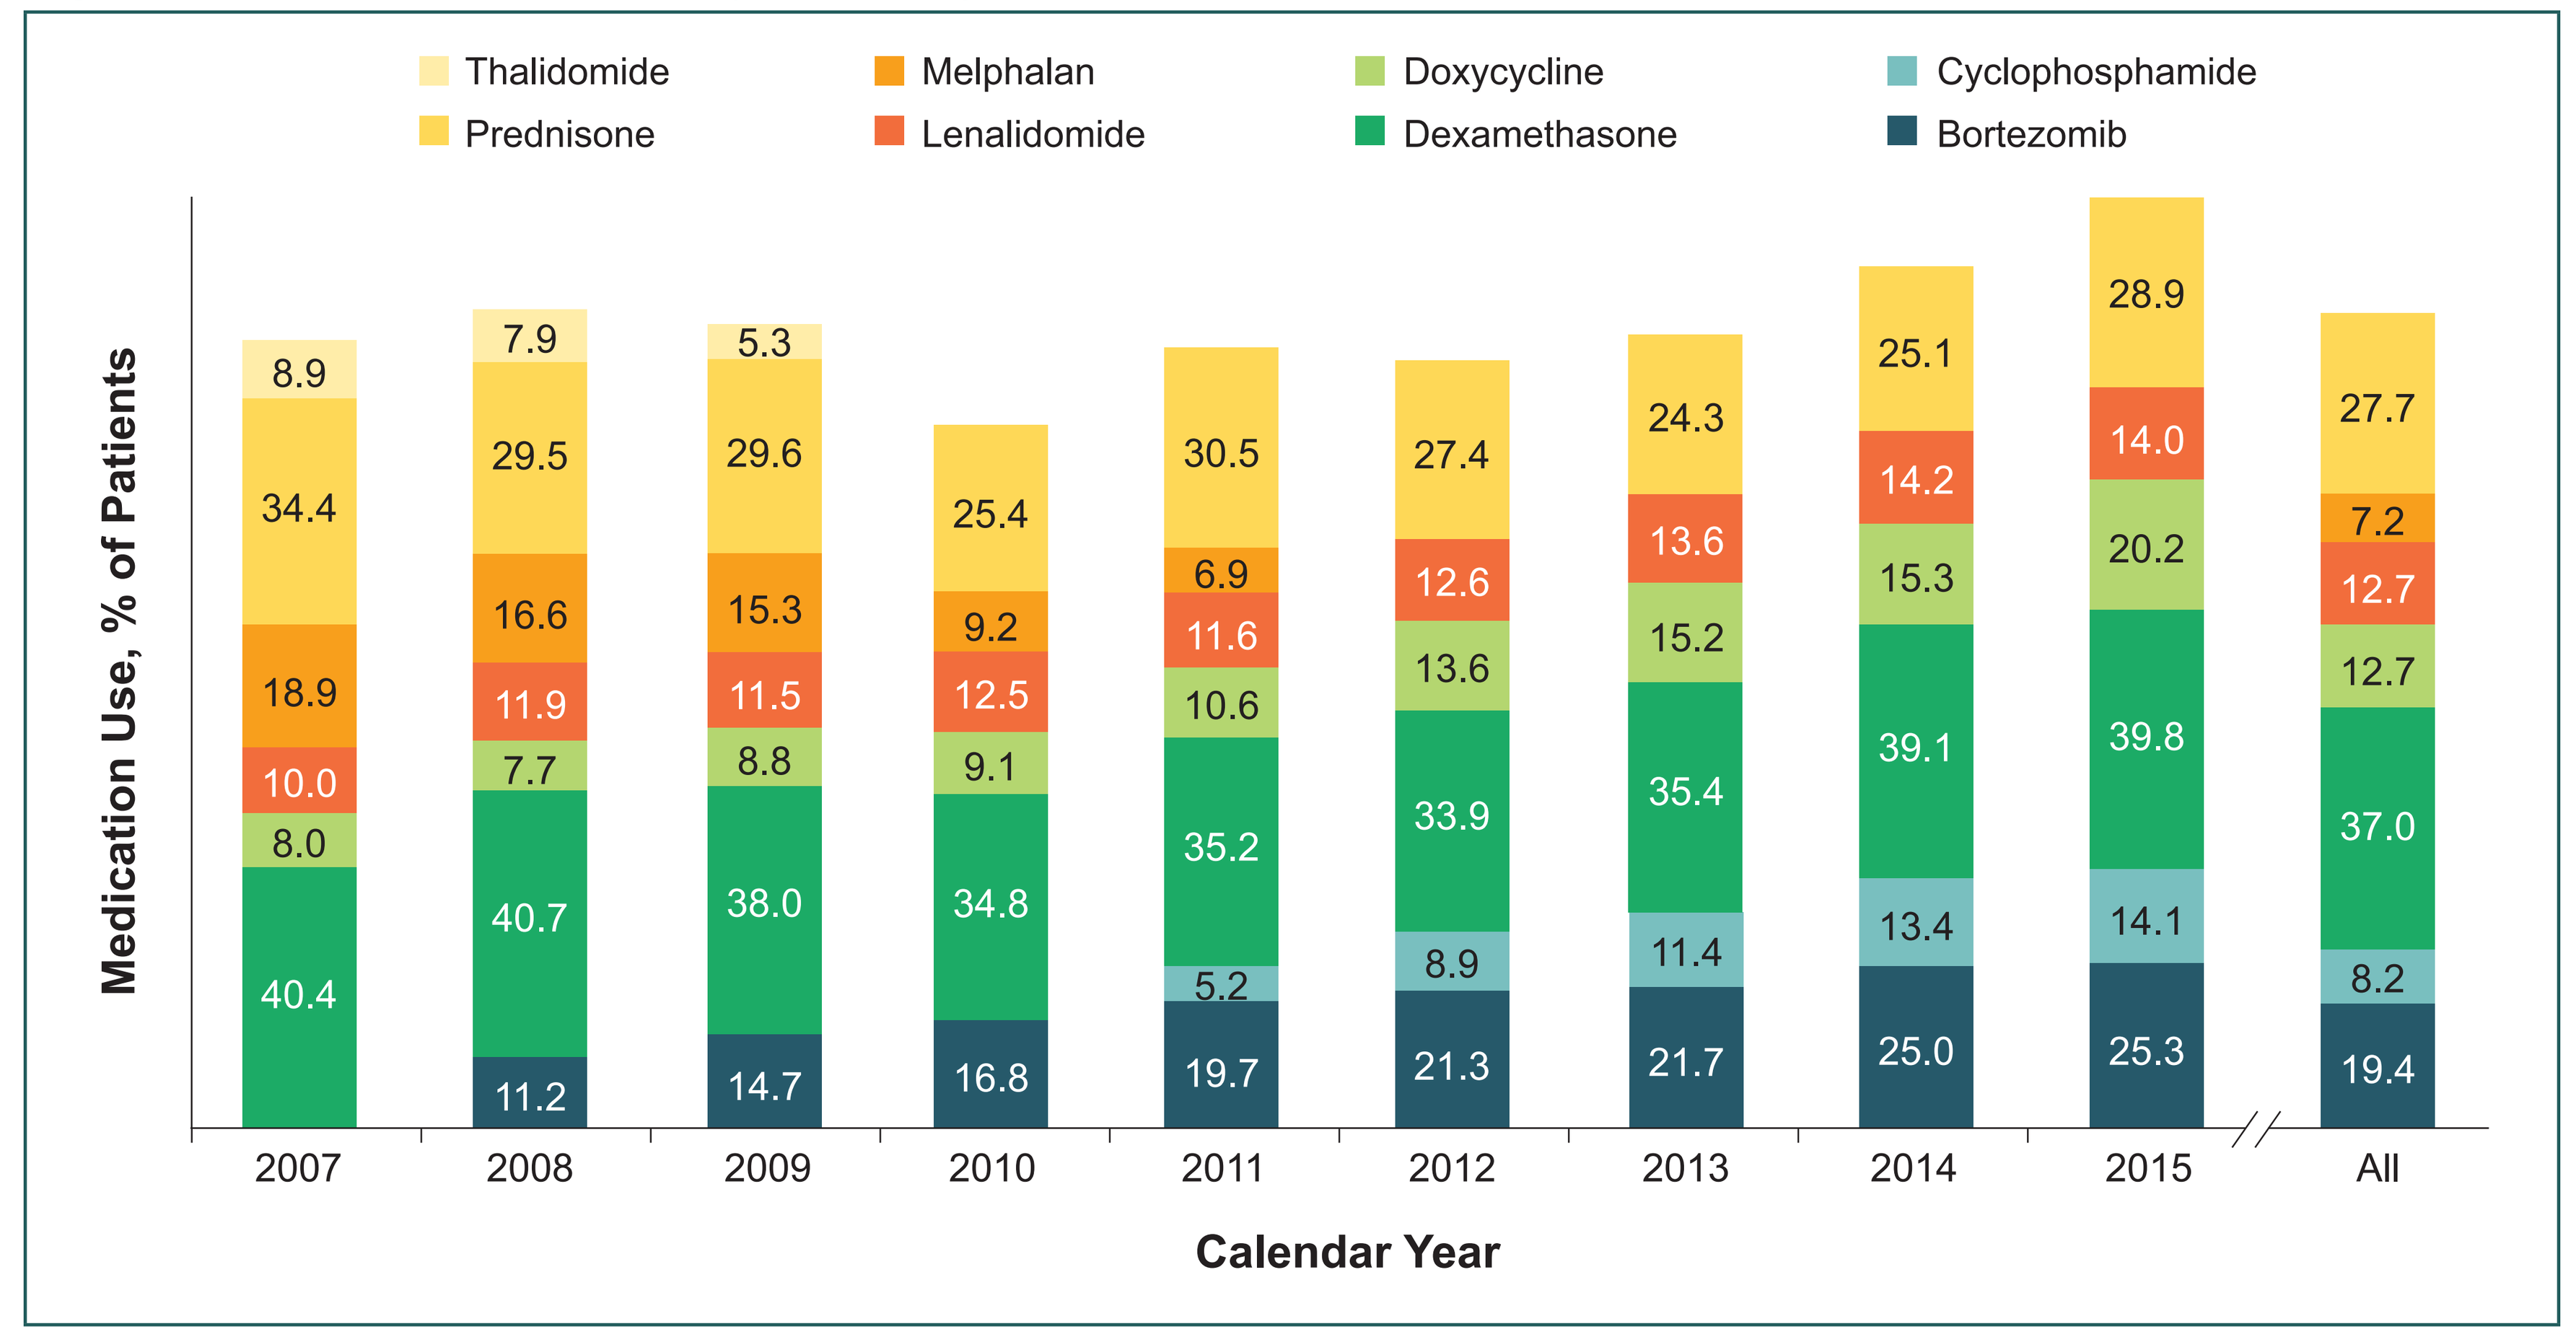
Thalidomide
Prednisone
Melphalan
Lenalidomide
Doxycycline
Dexamethasone
Cyclophosphamide
Bortezomib
Medication Use, % of Patients
2007
2008
2009
2010
2011
2012
2013
2014
2015
All
Calendar Year

[a]Percentages ≤5% are not shown.
[b]Patients could have received multiple treatments.

Service Utilization

- Hospital admission was common: 50.1% (n = 3670) of patients in the overall study sample were admitted ≥1 time, and 11.3% (n = 827) were admitted ≥3 times (**Table 2**)
- Among admitted patients, mean (±SD) length of stay was 14.7 (±19.5) days
 - Rates of any hospital admission decreased from 57.0% in 2007 to 46.2% in 2015
- More than one-third of all patients (34.3%; n = 2514) had ≥1 ED visit during the 9-year period, and 6.8% (n = 496) had ≥3 ED visits (**Table 2**). ED visits remained stable during the study period
- Patients had a mean (±SD) of 45.2 (±36.2) non–ED outpatient visits per year (**Table 2**). Patients were seen in an office setting a mean of 23.4 (±18.4) times per year. Non–ED outpatient visit rates increased during the study period from 30.0% in 2007 to 37.9% in 2015

Table 2. Inpatient and Outpatient Service Utilization by Calendar Year

Service	2007	2008	2009	2010	2011	2012	2013	2014	2015	All
N	349	573	713	792	972	1059	1031	1031	806	7326
Inpatient hospital admissions										
Mean (SD)	1.15 (1.45)	1.15 (1.49)	1.12 (1.59)	0.96 (1.42)	1.01 (1.54)	0.95 (1.44)	0.91 (1.30)	1.00 (1.51)	0.95 (1.76)	1.00 (1.50)
No. admissions, %										
0	43.0	43.3	45.7	50.5	50.7	50.4	51.7	52.2	53.8	49.9
1	25.5	28.3	27.8	26.5	25.6	27.2	26.1	23.0	24.2	25.9
2	18.9	14.1	13.6	12.2	11.4	12.6	12.3	12.7	12.8	12.9
≥3	12.6	14.3	12.9	10.7	12.2	9.8	9.9	12.1	9.2	11.3
Total days' stay, mean (SD)	n = 199 16.0 (20.2)	n = 325 15.4 (20.1)	n = 387 15.4 (22.7)	n = 392 14.0 (19.4)	n = 479 14.7 (17.8)	n = 525 12.9 (15.2)	n = 498 13.4 (18.0)	n = 493 16.0 (20.8)	n = 372 15.9 (22.3)	n = 3670 14.7 (19.5)
ED visits										
Mean (SD)	0.76 (1.98)	0.72 (2.19)	0.80 (2.11)	0.72 (2.55)	0.72 (2.17)	0.75 (2.81)	0.64 (1.63)	0.72 (1.41)	0.71 (1.65)	0.72 (2.10)
No. visits, %										
0	67.0	64.6	66.2	69.2	66.4	64.7	66.0	63.6	64.8	65.7
1	18.9	22.3	19.6	17.8	20.5	21.1	21.3	19.6	20.5	20.3
2	6.0	7.7	6.7	7.7	6.0	7.3	7.0	8.9	7.6	7.3
≥3	8.0	5.4	7.4	5.3	7.2	7.0	5.7	7.9	7.2	6.8
Non–ED outpatient services, mean (SD)	43.0 (30.0)	44.2 (37.9)	44.4 (37.7)	42.9 (35.2)	45.6 (39.0)	46.0 (35.4)	44.4 (32.8)	47.0 (36.8)	46.9 (37.9)	45.2 (36.2)
Office visits, mean (SD)	24.0 (16.5)	22.9 (17.5)	23.5 (19.3)	22.1 (17.6)	23.0 (18.3)	24.0 (18.2)	23.8 (18.3)	23.7 (19.0)	24.0 (19.7)	23.4 (18.4)
HSCT, %	4.0	3.3	4.8	3.5	3.0	3.7	3.3	4.7	3.6	3.7

ED, emergency department; HSCT, hematopoietic stem cell transplantation; SD, standard deviation.

Costs

- Mean (±SD) total annual all-cause healthcare costs were $101,855 (±$148,965) for all patients; most of the costs were for medical services
- Total costs increased during the study period, from $92,866 in 2007 to $114,030 in 2015, though pharmacy costs remained fairly stable (**Figure 2**)

Figure 2. Mean annual all-cause healthcare costs by calendar year.

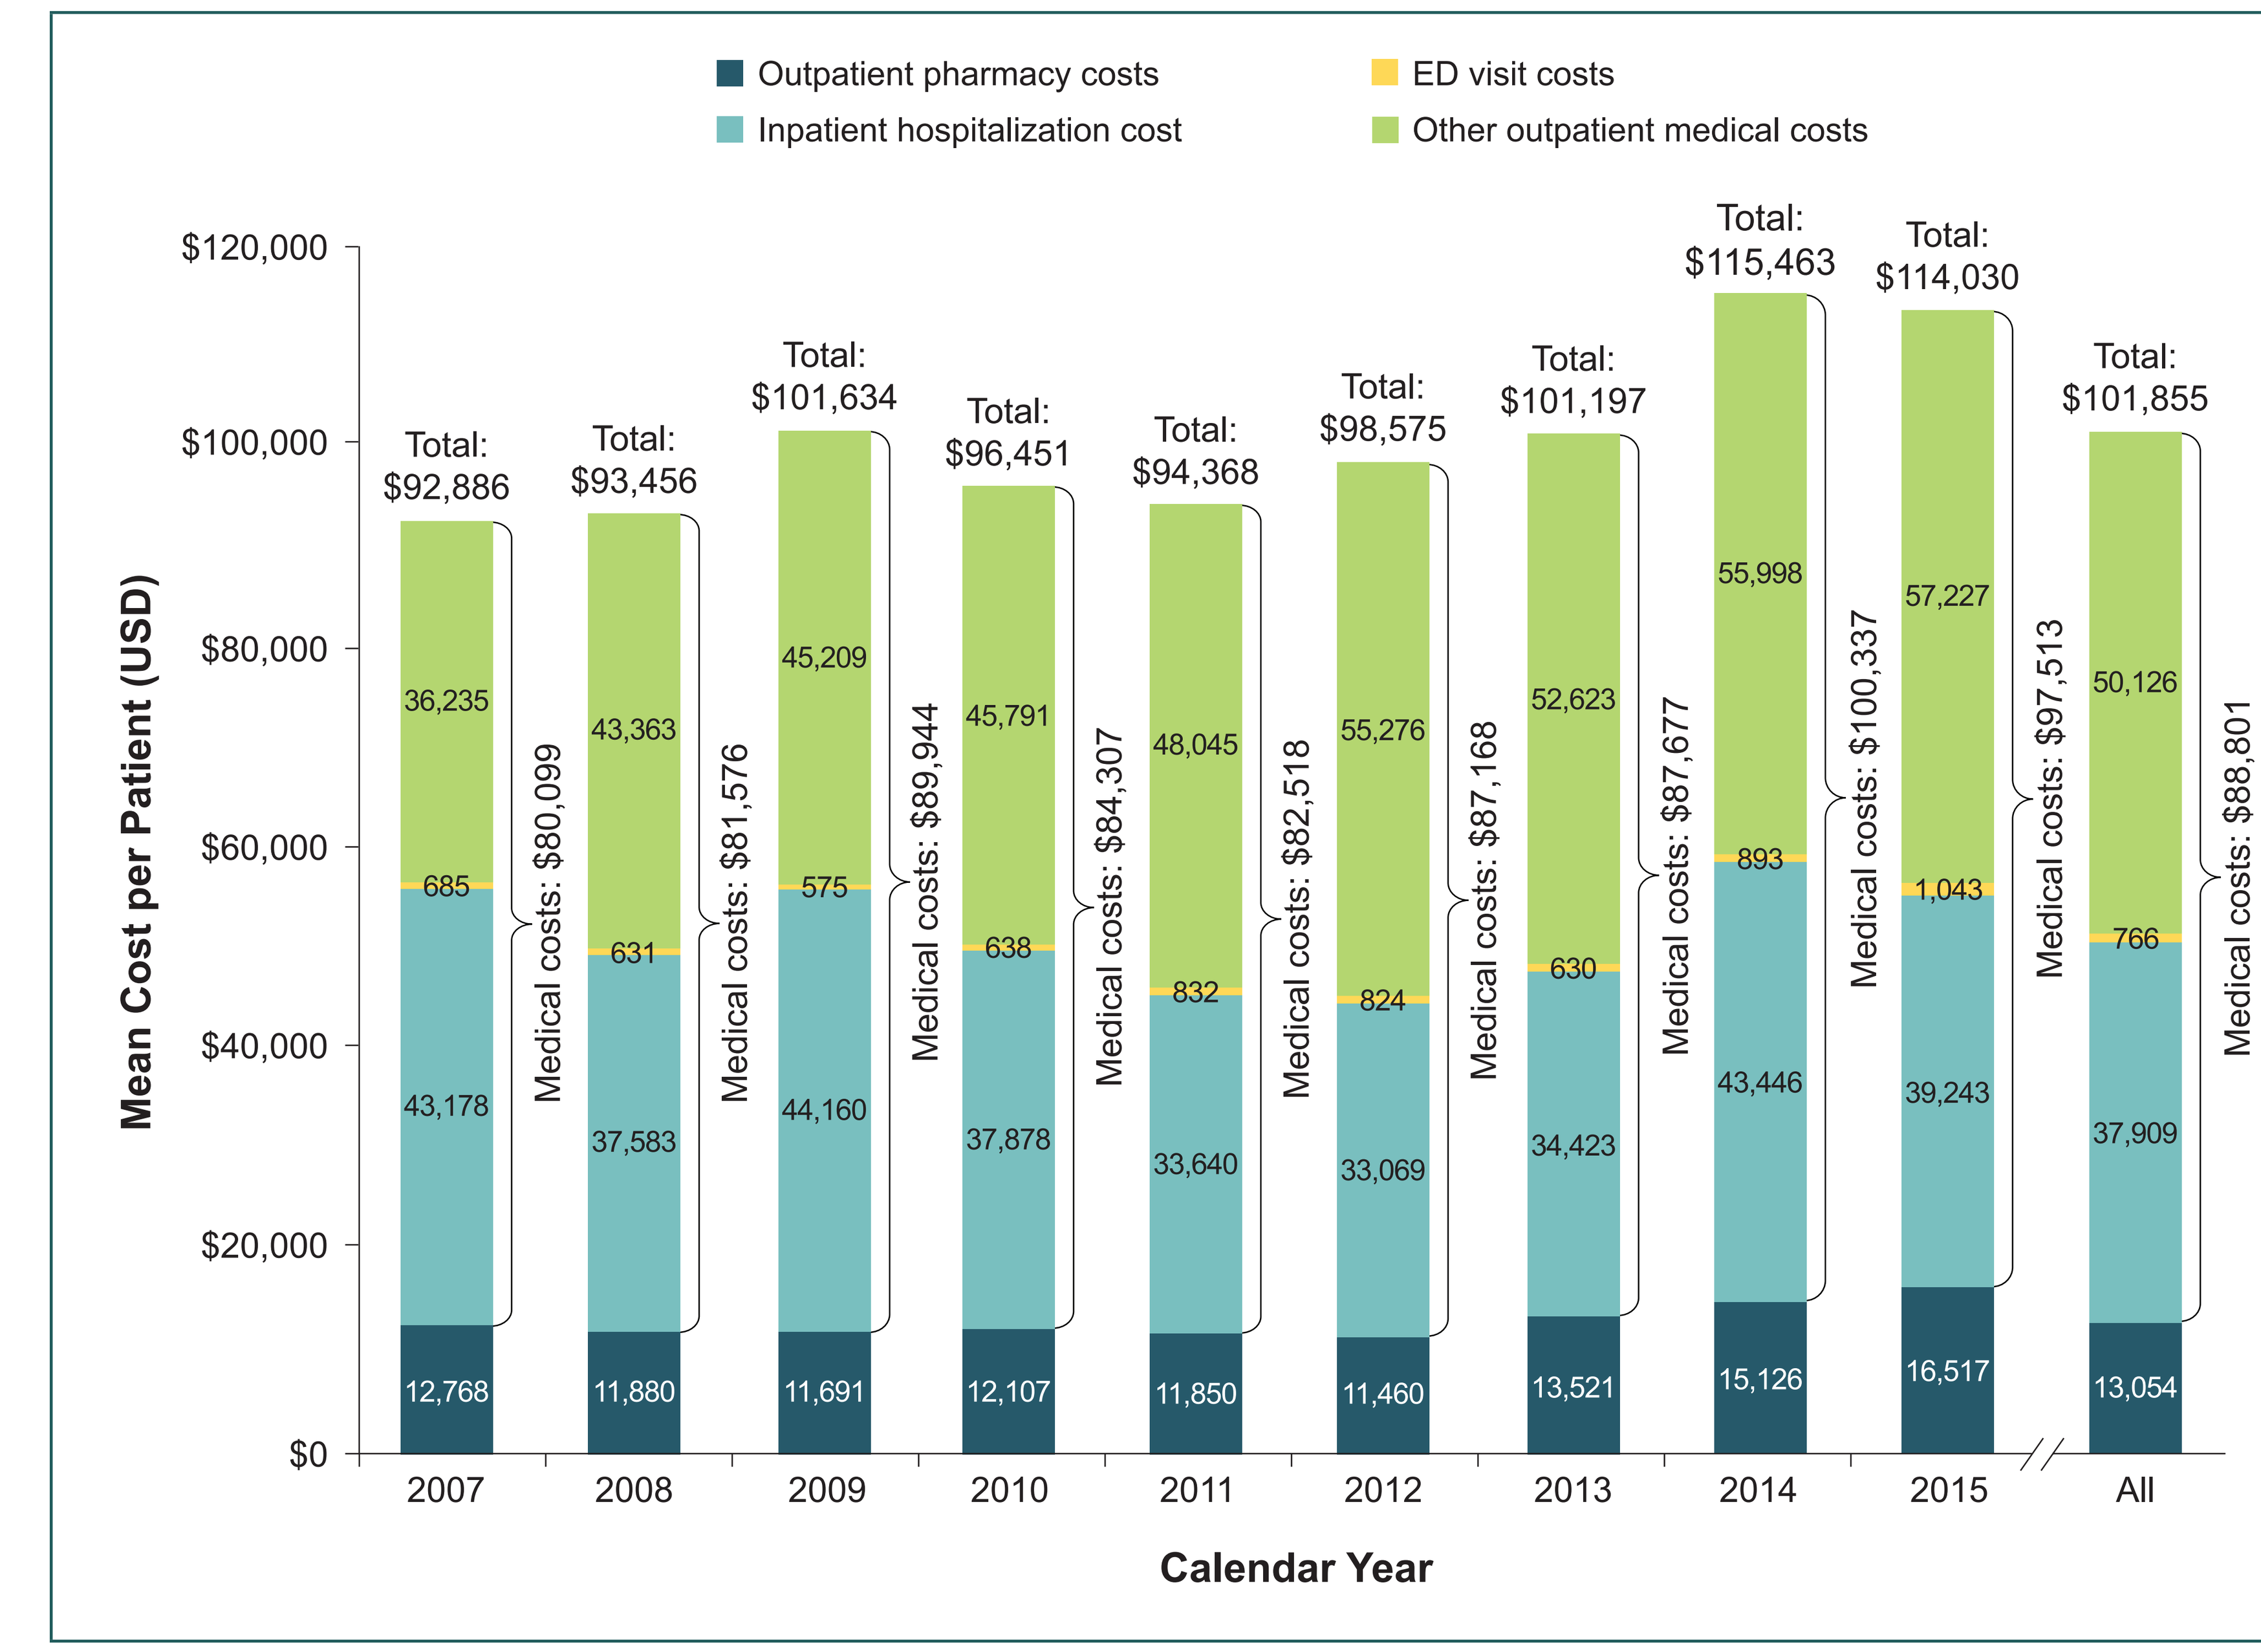
Outpatient pharmacy costs
Inpatient hospitalization cost
ED visit costs
Other outpatient medical costs
Mean Cost per Patient (USD)
Total: $92,886
Total: $93,456
Total: $101,634
Total: $96,451
Total: $94,368
Total: $98,575
Total: $101,197
Total: $115,463
Total: $114,030
Total: $101,855
Medical costs: $80,099
Medical costs: $81,576
Medical costs: $89,944
Medical costs: $84,307
Medical costs: $82,518
Medical costs: $87,168
Medical costs: $87,677
Medical costs: $100,337
Medical costs: $97,513
Medical costs: $88,801
Calendar Year

ED, emergency department; USD, United States dollars.

CONCLUSIONS

- The use of bortezomib-based regimens to treat patients with AL amyloidosis increased from 2007 to 2015
- Patients with AL amyloidosis use substantial healthcare resources
 - In a given year, half the patients with AL amyloidosis were admitted to the hospital at least once and had an average stay of >2 weeks
 - Visits to laboratories, offices, and other outpatient sites occurred almost twice a month
- The cost of this care totaled more than $100,000 per patient and rose during the period studied
- New therapies aimed at improving organ response have the potential to reduce disease burden and healthcare utilization

LIMITATIONS

- We identified AL amyloidosis patients with confidence given the algorithm we used; however, we acknowledge that existing *ICD-9-CM* and *ICD-10-CM* codes are not specific to AL amyloidosis, and we might have included a small number of ATTR amyloidosis patients in this study
- Approximately 38.9% of our study population had coexisting multiple myeloma, based on having ≥1 claim with a relevant *ICD-9-CM* or *ICD-10-CM* code. Our data are not clinically detailed enough to determine which diagnosis was most appropriate for these patients
- Our data source represents care for persons (or their dependents) who are healthy enough to work and have commercial or Medicare Supplement Insurance. Given this healthy-worker bias, utilization and costs reported here may be underestimated
- Our cost estimates include direct healthcare costs only and do not take into account important indirect costs associated with caregiver burden, loss of productivity, and reduced quality of life

REFERENCES

1. Mahmood S et al. *Haematologica*. 2014;99(2):209-221.
2. Merlini G, Bellotti V. *N Engl J Med*. 2003;349(6):583-596.
3. Sanchorawala V. *Clin J Am Soc Nephrol*. 2006;1(6):1331-1341.
4. Lin HM et al. *CMRO*. 2017;33(6):1017-1031.

Disclosures of Interest

TPQ is a full-time employee of Prothena Biosciences Inc and a stockholder in Prothena Corporation plc. JTY, EC, and MSB are full-time employees of Partnership for Health Analytic Research, LLC. SDG has nothing to disclose.

http://bit.ly/2A0CLgV

Presented at the 59th ASH Annual Meeting & Exposition; December 9–12, 2017; Atlanta, Georgia