Real-world Treatment Utilization of Sodium Oxybate in Adult Patients With Narcolepsy: An Analysis of Claims Data

Annual Meeting of the Academy of Managed Care Pharmacy (AMCP) Nexus
October 29-November 1, 2019 • National Harbor, MD

Aatif M. Husain, MD[1]; Chad M. Ruoff, MD[2]; Sheila Reiss Reddy, PhD, RPh[3]; Ryan S. Tieu, MS[3]; Danielle Hyman, PhD[4]; Judith Profant, PhD, DBSM[4]; Mo Yang, PhD[4]; Morgan Bron, PharmD, MS[5]; Kathleen F. Villa, MS[5]

[1]Duke University Medical Center, Duke Clinical Research Institute, and Veterans Affairs Medical Center, Durham, NC, USA; [2]Stanford Center for Sleep Sciences and Medicine, Palo Alto, CA, USA; [3]Partnership for Health Analytic Research, LLC, Beverly Hills, CA, USA; [4]Jazz Pharmaceuticals, Inc., Palo Alto, CA, USA; [5]Former employee of Jazz Pharmaceuticals, Inc., Palo Alto, CA, USA.

Background

- Narcolepsy is a rare, lifelong, neurological disorder characterized by excessive daytime sleepiness and dysregulation of the sleep-wake cycle, including cataplexy, hypnagogic hallucinations, and sleep paralysis, though not all patients experience all symptoms[1]
- Sodium oxybate (SXB) is considered a standard of care for the treatment of narcolepsy[2]
- In narcolepsy studies, rates of SXB treatment compliance and adherence were generally high[3-5]
 - However, real-world SXB utilization has not been well studied

Objective

- To examine SXB adherence, persistence, and overall treatment utilization patterns in real-world settings in adult patients with narcolepsy in the United States

Methods

- This retrospective cohort study identified US patients ≥18 years of age diagnosed with narcolepsy using de-identified commercial insurance claims from the Truven Health Analytics MarketScan® Commercial Claims and Encounters database from October 2012 through September 2017
- Patients with narcolepsy were first identified based on having ≥1 of the following: ≥1 nondiagnostic claim with a code for narcolepsy with or without cataplexy and ≥1 claim for SXB; ≥2 claims with a code for narcolepsy (1 being nondiagnostic); or ≥1 nondiagnostic claim with a code for narcolepsy following a Multiple Sleep Latency Test (MSLT)
 - Nondiagnostic claims were defined as claims for evaluation and management services without MSLT/polysomnography or other diagnostic testing
- For the analysis of SXB treatment patterns, narcolepsy patients who were being treated with ≥1 claim for SXB during the identification (ID) period (October 1, 2013-September 30, 2016) were identified
 - The first claim for SXB during the ID period was defined as the index date
 - Patients were required to have continuous enrollment for ≥1 year prior to and ≥1 year after the index date, with ≥1 nondiagnostic claim prior to the index date
 - New users were defined as those who did not have any SXB claims during the 1-year period prior to the index date; continuing users were defined as those who had ≥1 SXB claim during the 1-year period prior to the index date

Figure 1. Study Timeline for New Versus Continuing SXB Users

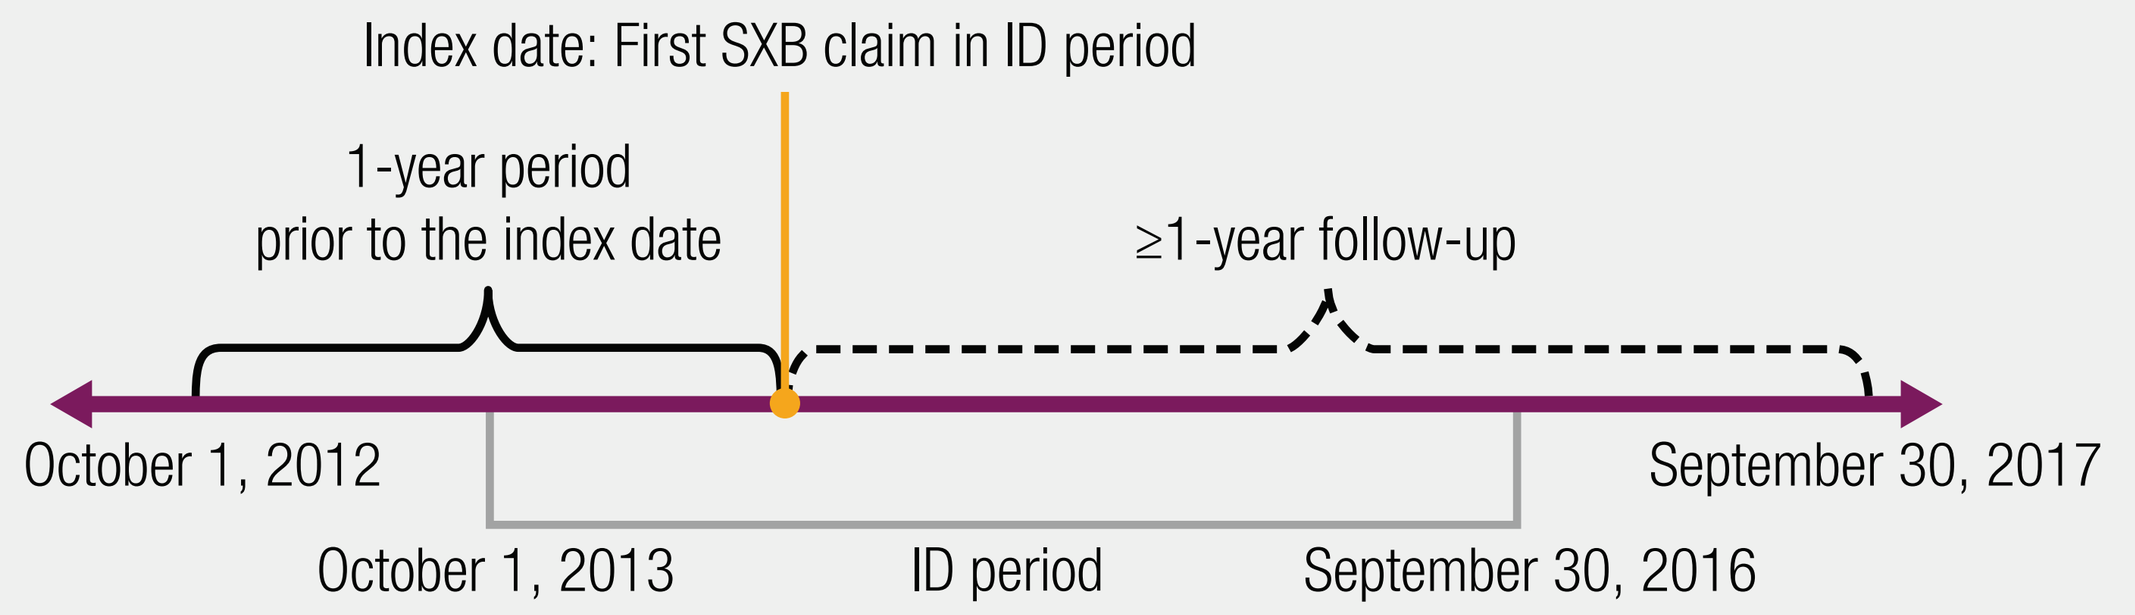
Index date: First SXB claim in ID period
1-year period
prior to the index date
≥1-year follow-up
October 1, 2012
September 30, 2017
October 1, 2013
ID period
September 30, 2016

SXB, sodium oxybate; ID, identification.

- Treatment utilization, adherence, and persistence of SXB were measured at 3 months, 6 months, 9 months, 1 year, and 2 years
 - Adherence was assessed as mean proportion of days covered (PDC) and proportion of patients with PDC ≥80%, and persistence was evaluated by mean number of days on continuous treatment (defined as treatment with an interruption of <60 days)
- Descriptive statistics were reported, and chi-square or *t* tests were performed to compare between-group treatment patterns. Kaplan-Meier curves were produced, and log-rank tests were performed to compare continuous time on treatment. *P* values were not controlled for multiplicity and, hence, are nominal
- Graphical analyses were performed to examine overall treatment patterns among adults with narcolepsy treated with any narcolepsy-related treatments (both indicated medications and those used to treat common comorbid conditions in patients with narcolepsy), including SXB, wake-promoting agents (WPAs), attention-deficit/hyperactivity disorder (ADHD) agents and traditional central nervous system stimulants, antidepressants, and nonstimulant ADHD agents
 - The first claim for any narcolepsy-related treatment was considered the index date, with similar continuous enrollment requirements as described above
 - Patients were stratified as treatment-naïve (no treatment prior to index), newly treated with index treatment (no index treatment/treatment combination prior to index but who were not treatment-naïve), or continued index treatment (index treatment/treatment combination prior to index)

Results

Table 1. Demographics of Adult Narcolepsy Patients Treated With SXB

Characteristic	All (n = 1,280)	New users (n = 633)	Continuing users (n = 647)	*P* value
Age, years, mean (SD)	39.3 (13.6)	36.8 (12.8)	41.8 (13.8)	<0.001
Age group, n (%)				<0.001
18-24 years	246 (19.2)	152 (24.0)	94 (14.5)	
25-44 years	584 (45.6)	302 (47.7)	282 (43.6)	
45-64 years	410 (32.0)	173 (27.3)	237 (36.6)	
≥65 years	40 (3.1)	6 (0.9)	34 (5.3)	
Female, n (%)	865 (67.6)	439 (69.4)	426 (65.8)	0.179
Geographic region, n (%)				0.025
Midwest	345 (27.0)	149 (23.5)	196 (30.3)	
Northeast	156 (12.2)	86 (13.6)	70 (10.8)	
South	623 (48.7)	324 (51.2)	299 (46.2)	
West	156 (12.2)	74 (11.7)	82 (12.7)	
Insurance type, n (%)				0.178
Comprehensive	49 (3.8)	19 (3.0)	30 (4.6)	
EPO	9 (0.7)	4 (0.6)	5 (0.8)	
HMO	114 (8.9)	51 (8.1)	63 (9.7)	
POS	82 (6.4)	36 (5.7)	46 (7.1)	
PPO	756 (59.1)	387 (61.1)	369 (57.0)	
POS with capitation	9 (0.7)	3 (0.5)	6 (0.9)	
CDHP	133 (10.4)	76 (12.0)	57 (8.8)	
HDHP	113 (8.8)	52 (8.2)	61 (9.4)	
Missing/unknown	15 (1.2)	5 (0.8)	10 (1.5)	

SXB, sodium oxybate; SD, standard deviation; EPO, exclusive provider organization; HMO, health maintenance organization; POS, point of service; PPO, preferred provider organization; CDHP, consumer-directed health plan; HDHP, high-deductible health plan.

- Among 1,280 continuously enrolled patients taking SXB (mean [±standard deviation] age, 39.3 [±13.6] years; 67.6% female), 633 were new SXB users and 647 were continuing SXB users

Figure 2. Initial Treatment Regimen[a,b] of Adult Narcolepsy Patients Newly Treated With SXB (n = 633)

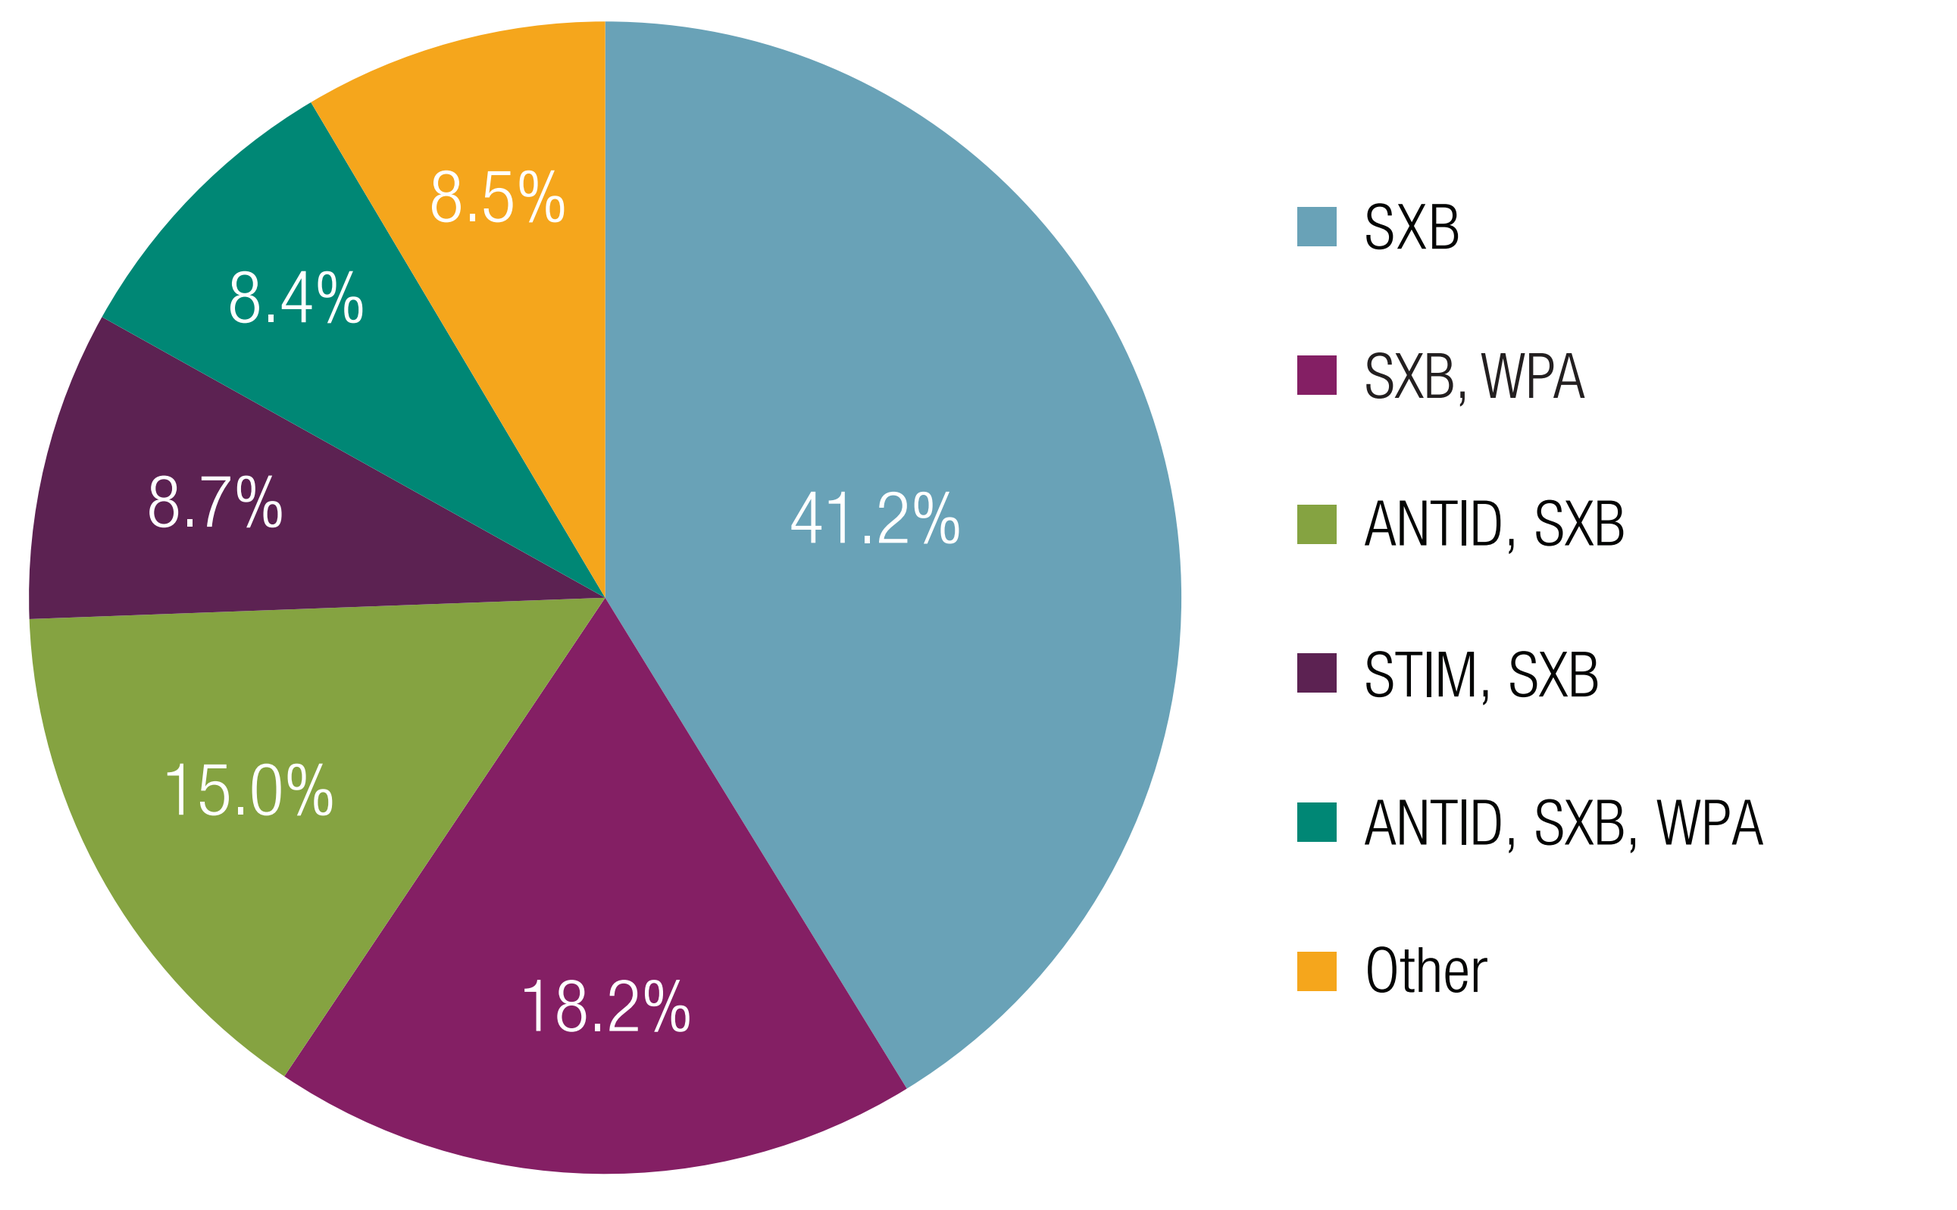
8.5%
8.4%
8.7%
41.2%
15.0%
18.2%
SXB
SXB, WPA
ANTID, SXB
STIM, SXB
ANTID, SXB, WPA
Other

'Other' included:	Percentage
STIM, ANTID, SXB	4.3
STIM, SXB, WPA	2.2
STIM, ANTID, SXB, WPA	1.0
ADHD, ANTID, SXB	0.5
ADHD, SXB	0.3
ADHD, STIM, ANTID, SXB, WPA	0.2
ADHD, SXB, WPA	0.2

SXB, sodium oxybate; WPA, wake-promoting agent; ANTID, antidepressant; STIM, stimulant; ADHD, attention-deficit/hyperactivity disorder; CNS, central nervous system.
[a]Defined as all treatment classes occurring between the index date and 45 days after the index date.
[b]Regarding treatment classes, ADHD represents nonstimulant ADHD agents, while STIM represents traditional CNS stimulants (eg, amphetamine, methylphenidate).

- Among 633 new users, 41.2% (n = 261) received monotherapy and 58.8% (n = 372) received combination therapy
- Of new users receiving combination therapy (n = 372), 30.9% (n = 115) received SXB plus a WPA, and 25.5% (n = 95) received SXB plus an antidepressant

Figure 3. (A) Mean PDC for New and Continuing SXB Users and (B) Proportion of New and Continuing SXB Users With PDC ≥80%

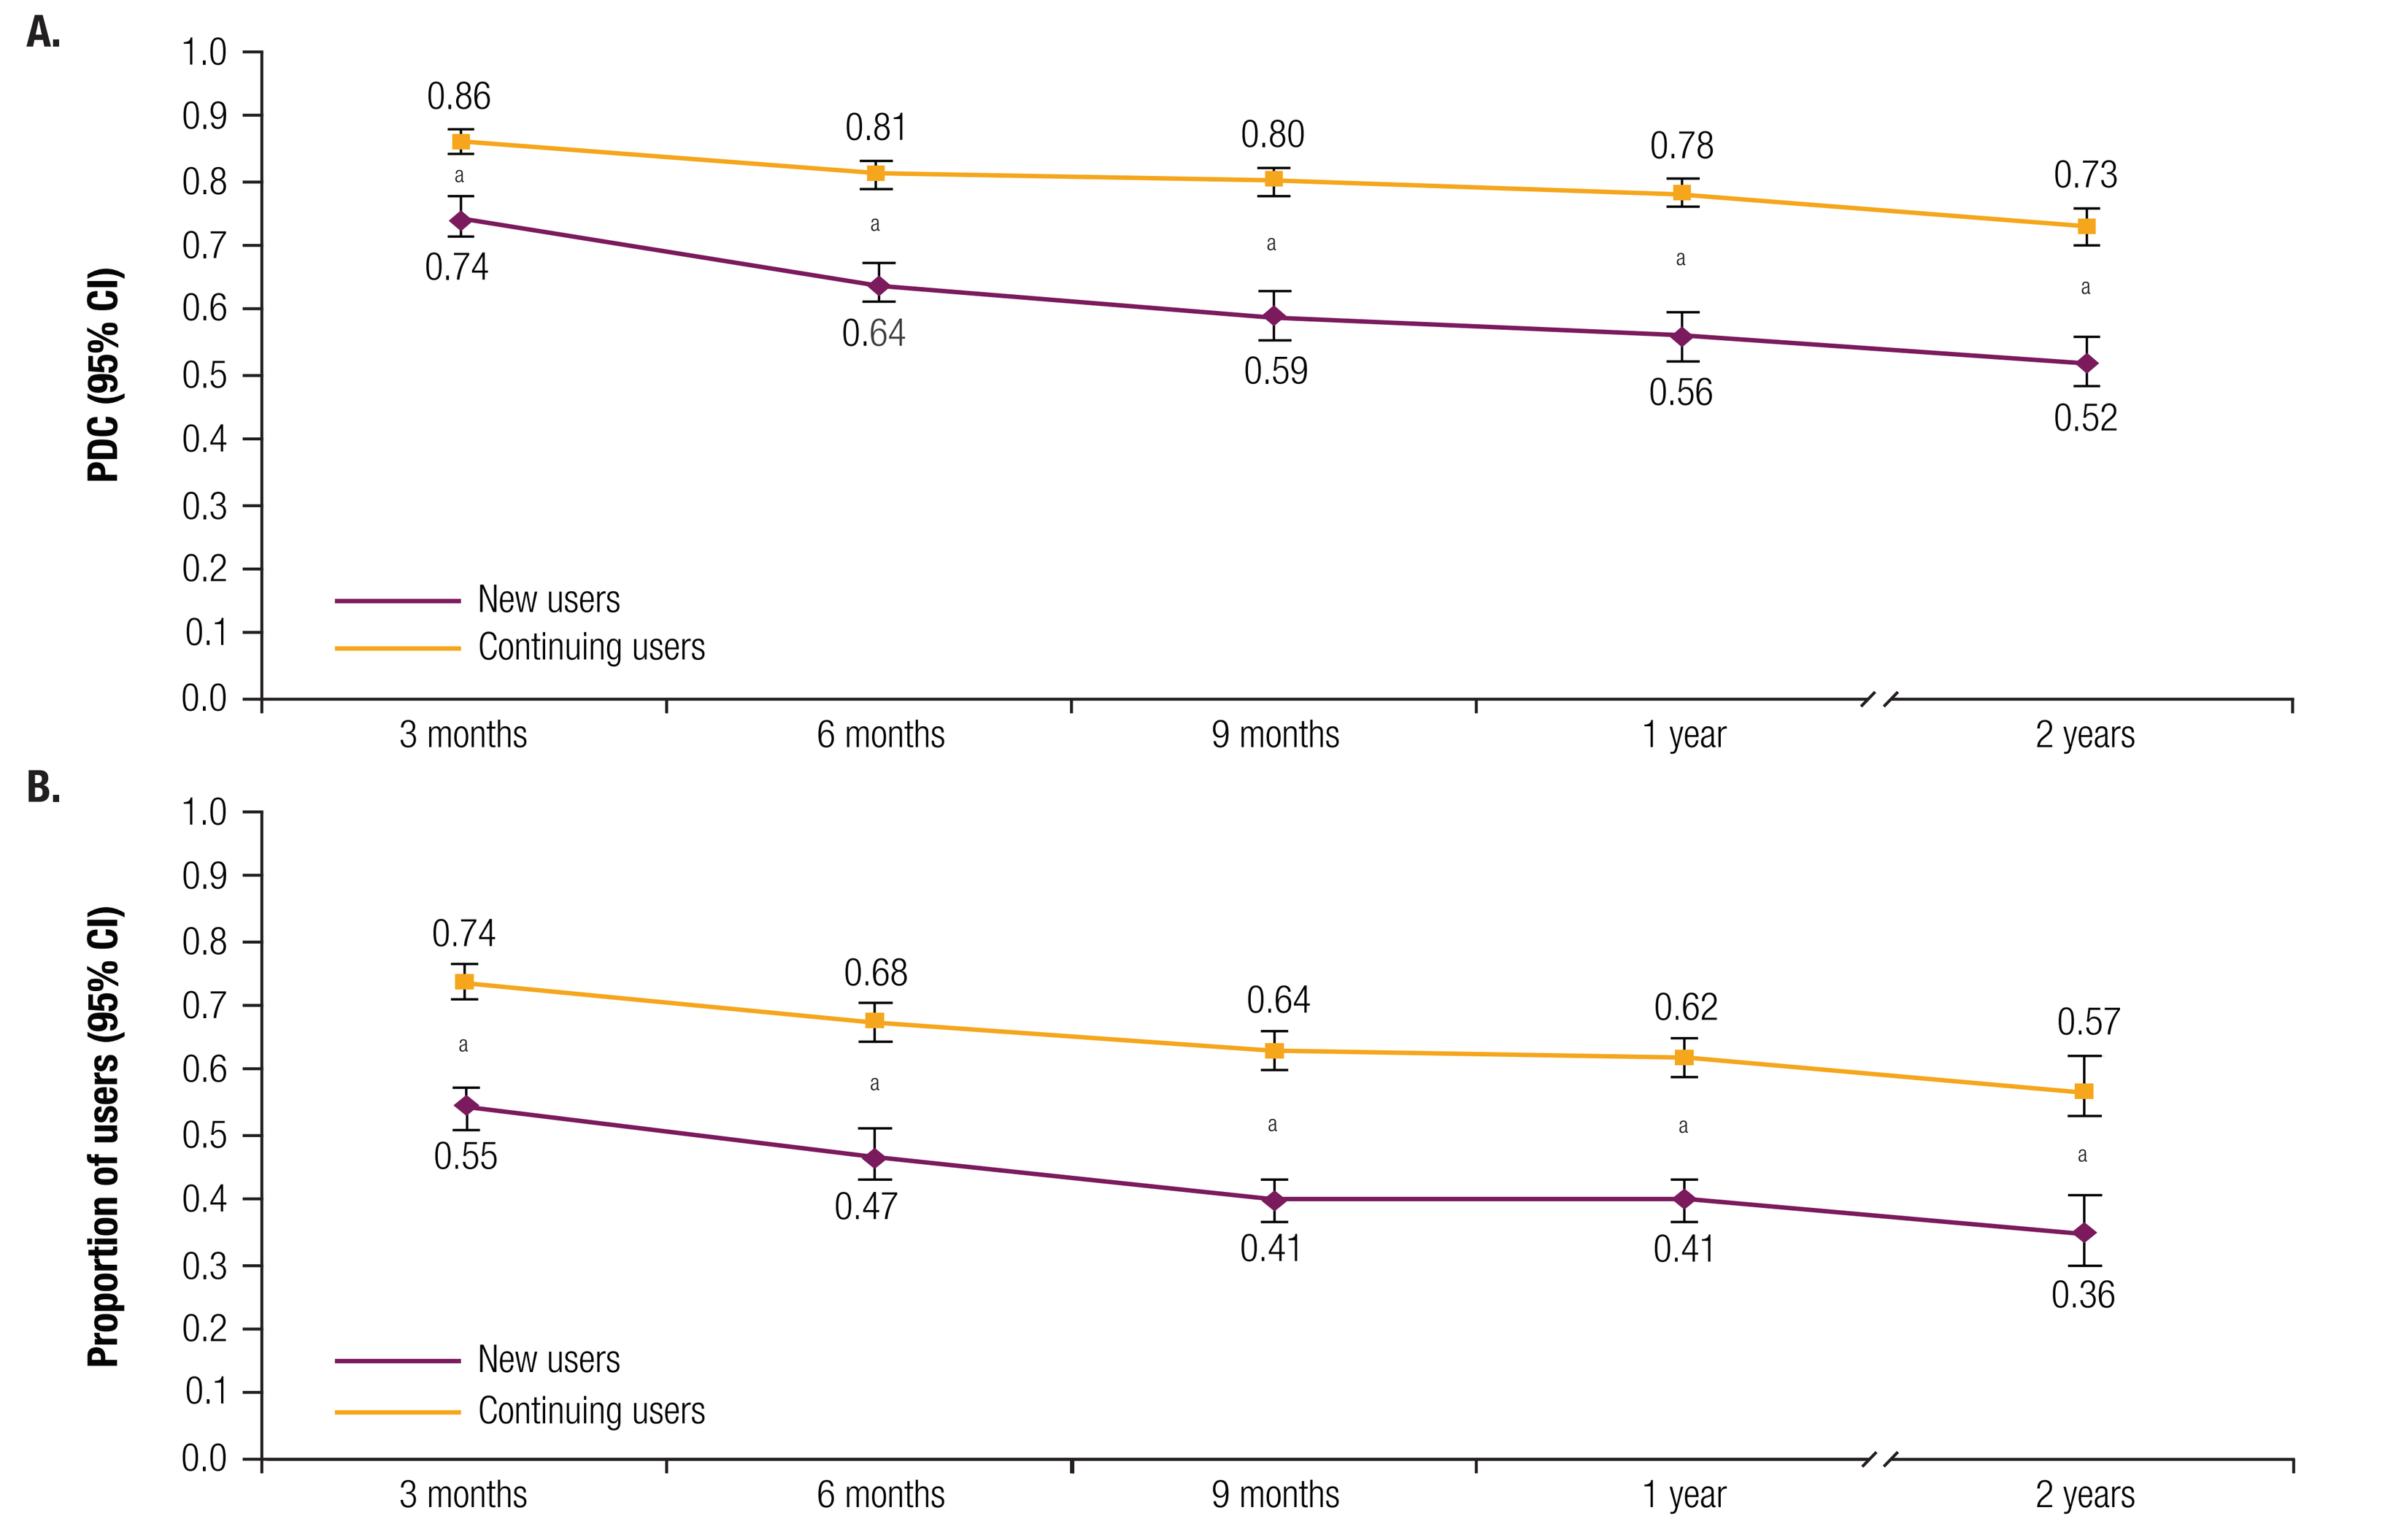
A.
PDC (95% CI)
0.86
0.81
0.80
0.78
0.73
0.74
0.64
0.59
0.56
0.52
New users
Continuing users
3 months
6 months
9 months
1 year
2 years
B.
Proportion of users (95% CI)
0.74
0.68
0.64
0.62
0.57
0.55
0.47
0.41
0.41
0.36
New users
Continuing users
3 months
6 months
9 months
1 year
2 years

PDC, proportion of days covered; SXB, sodium oxybate; CI, confidence interval.
[a]$P<0.001$.

- Mean PDC and PDC ≥80% decreased over time for new users; for continuing users, mean PDC remained relatively high and constant, and PDC ≥80% decreased over time

Figure 4. Time to Discontinuation of SXB for New and Continuing Users

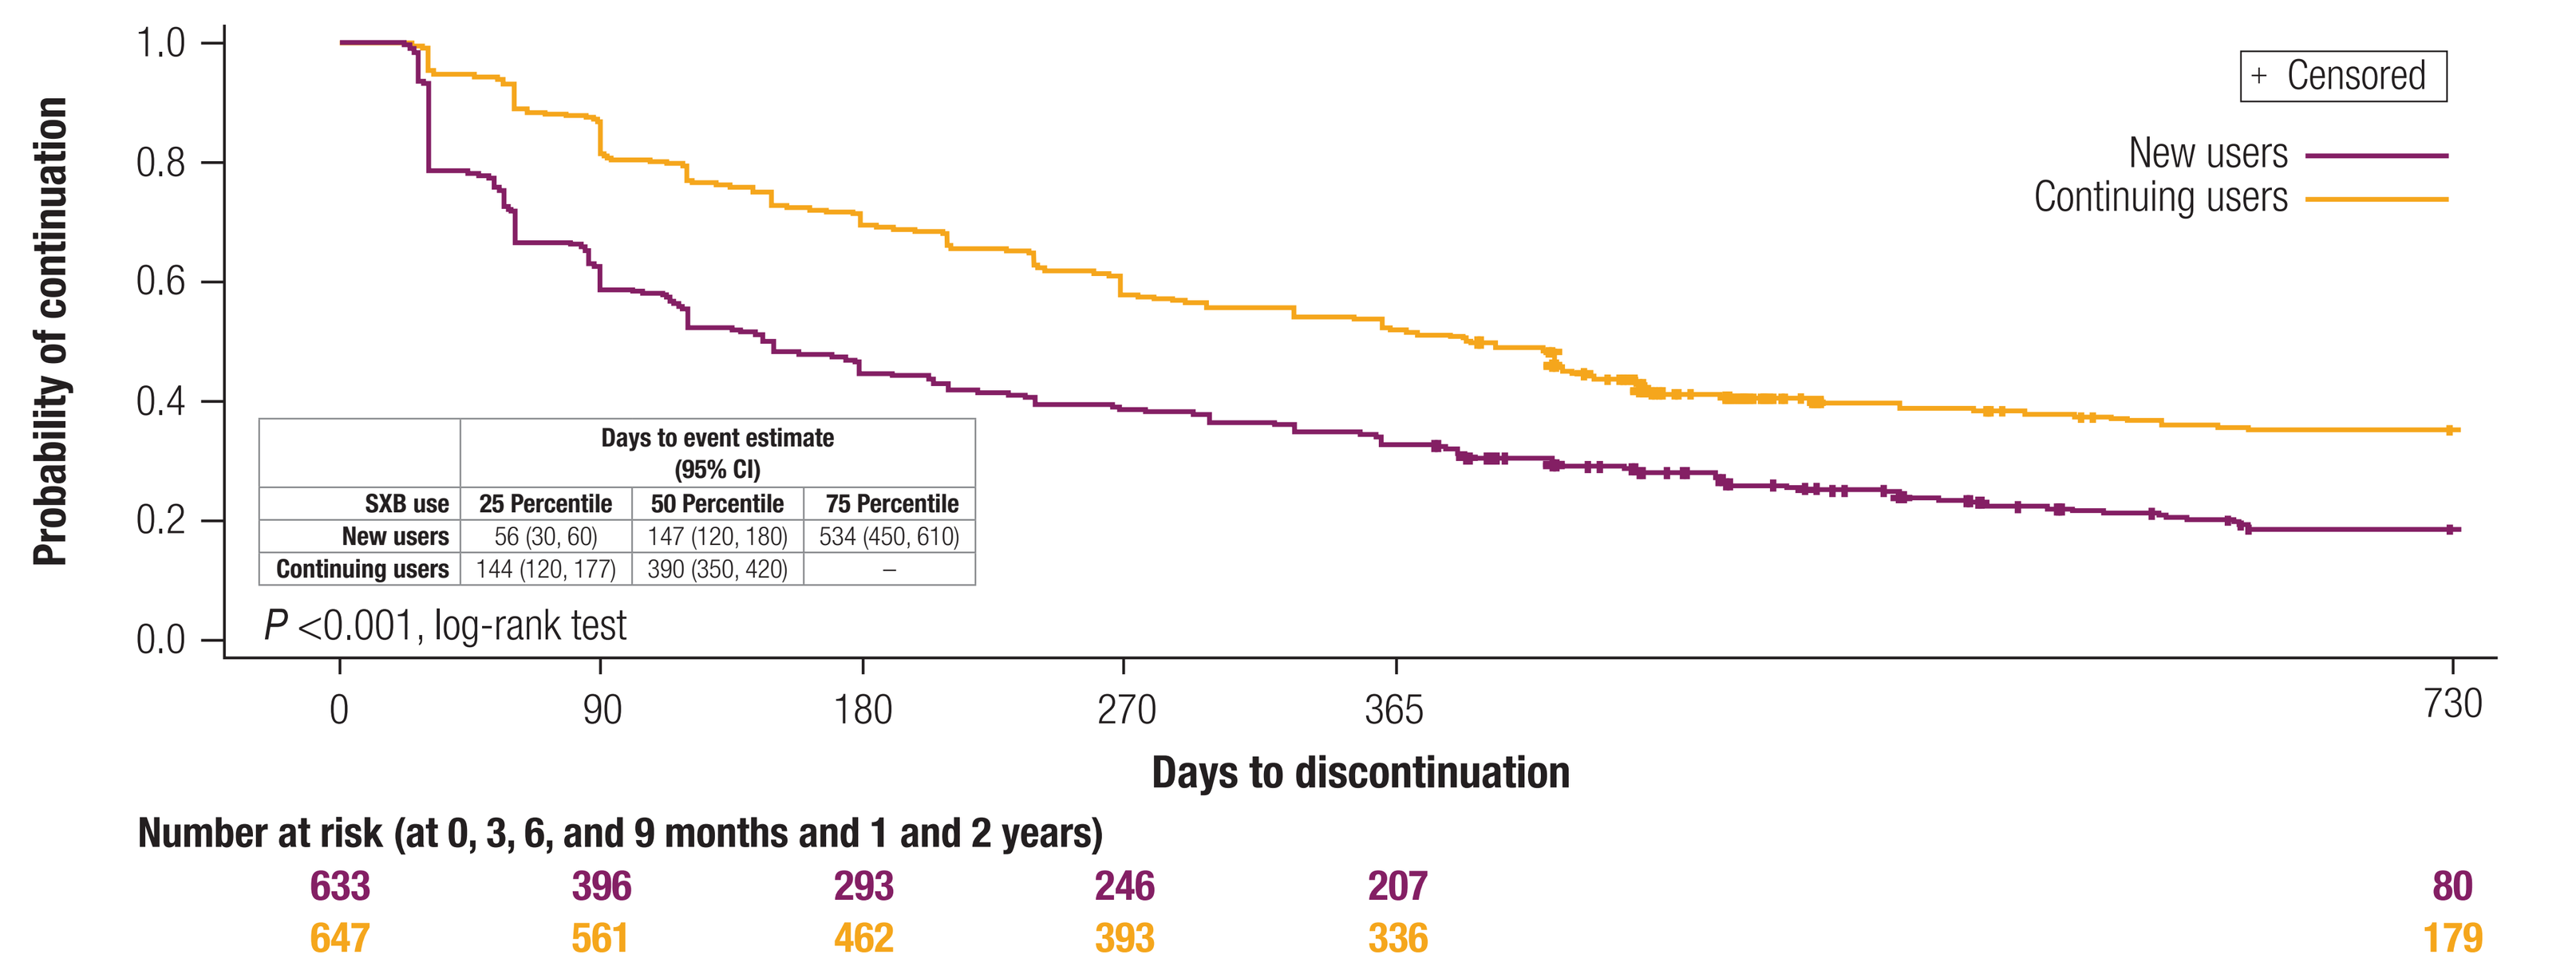
+ Censored
New users
Continuing users
Probability of continuation
P <0.001, log-rank test
0
90
180
270
365
730
Days to discontinuation
Number at risk (at 0, 3, 6, and 9 months and 1 and 2 years)
633 396 293 246 207 80
647 561 462 393 336 179

SXB use	Days to event estimate (95% CI): 25 Percentile	50 Percentile	75 Percentile
New users	56 (30, 60)	147 (120, 180)	534 (450, 610)
Continuing users	144 (120, 177)	390 (350, 420)	–

SXB, sodium oxybate; CI, confidence interval.

- The median (95% confidence interval) time to SXB discontinuation was 147 (120-180) days for new users and 390 (350-420) days for continuing users
 - Differences were observed in the number of days to discontinuation for new versus continuing SXB users (nominal *P* <0.001, log-rank test)

Figure 5. GRAPHx Patterns of Narcolepsy-related Treatment for Patients Who Were Treatment-naïve Prior to Index, Were Newly Treated With Index Treatment, and Continued Index Treatment[a]

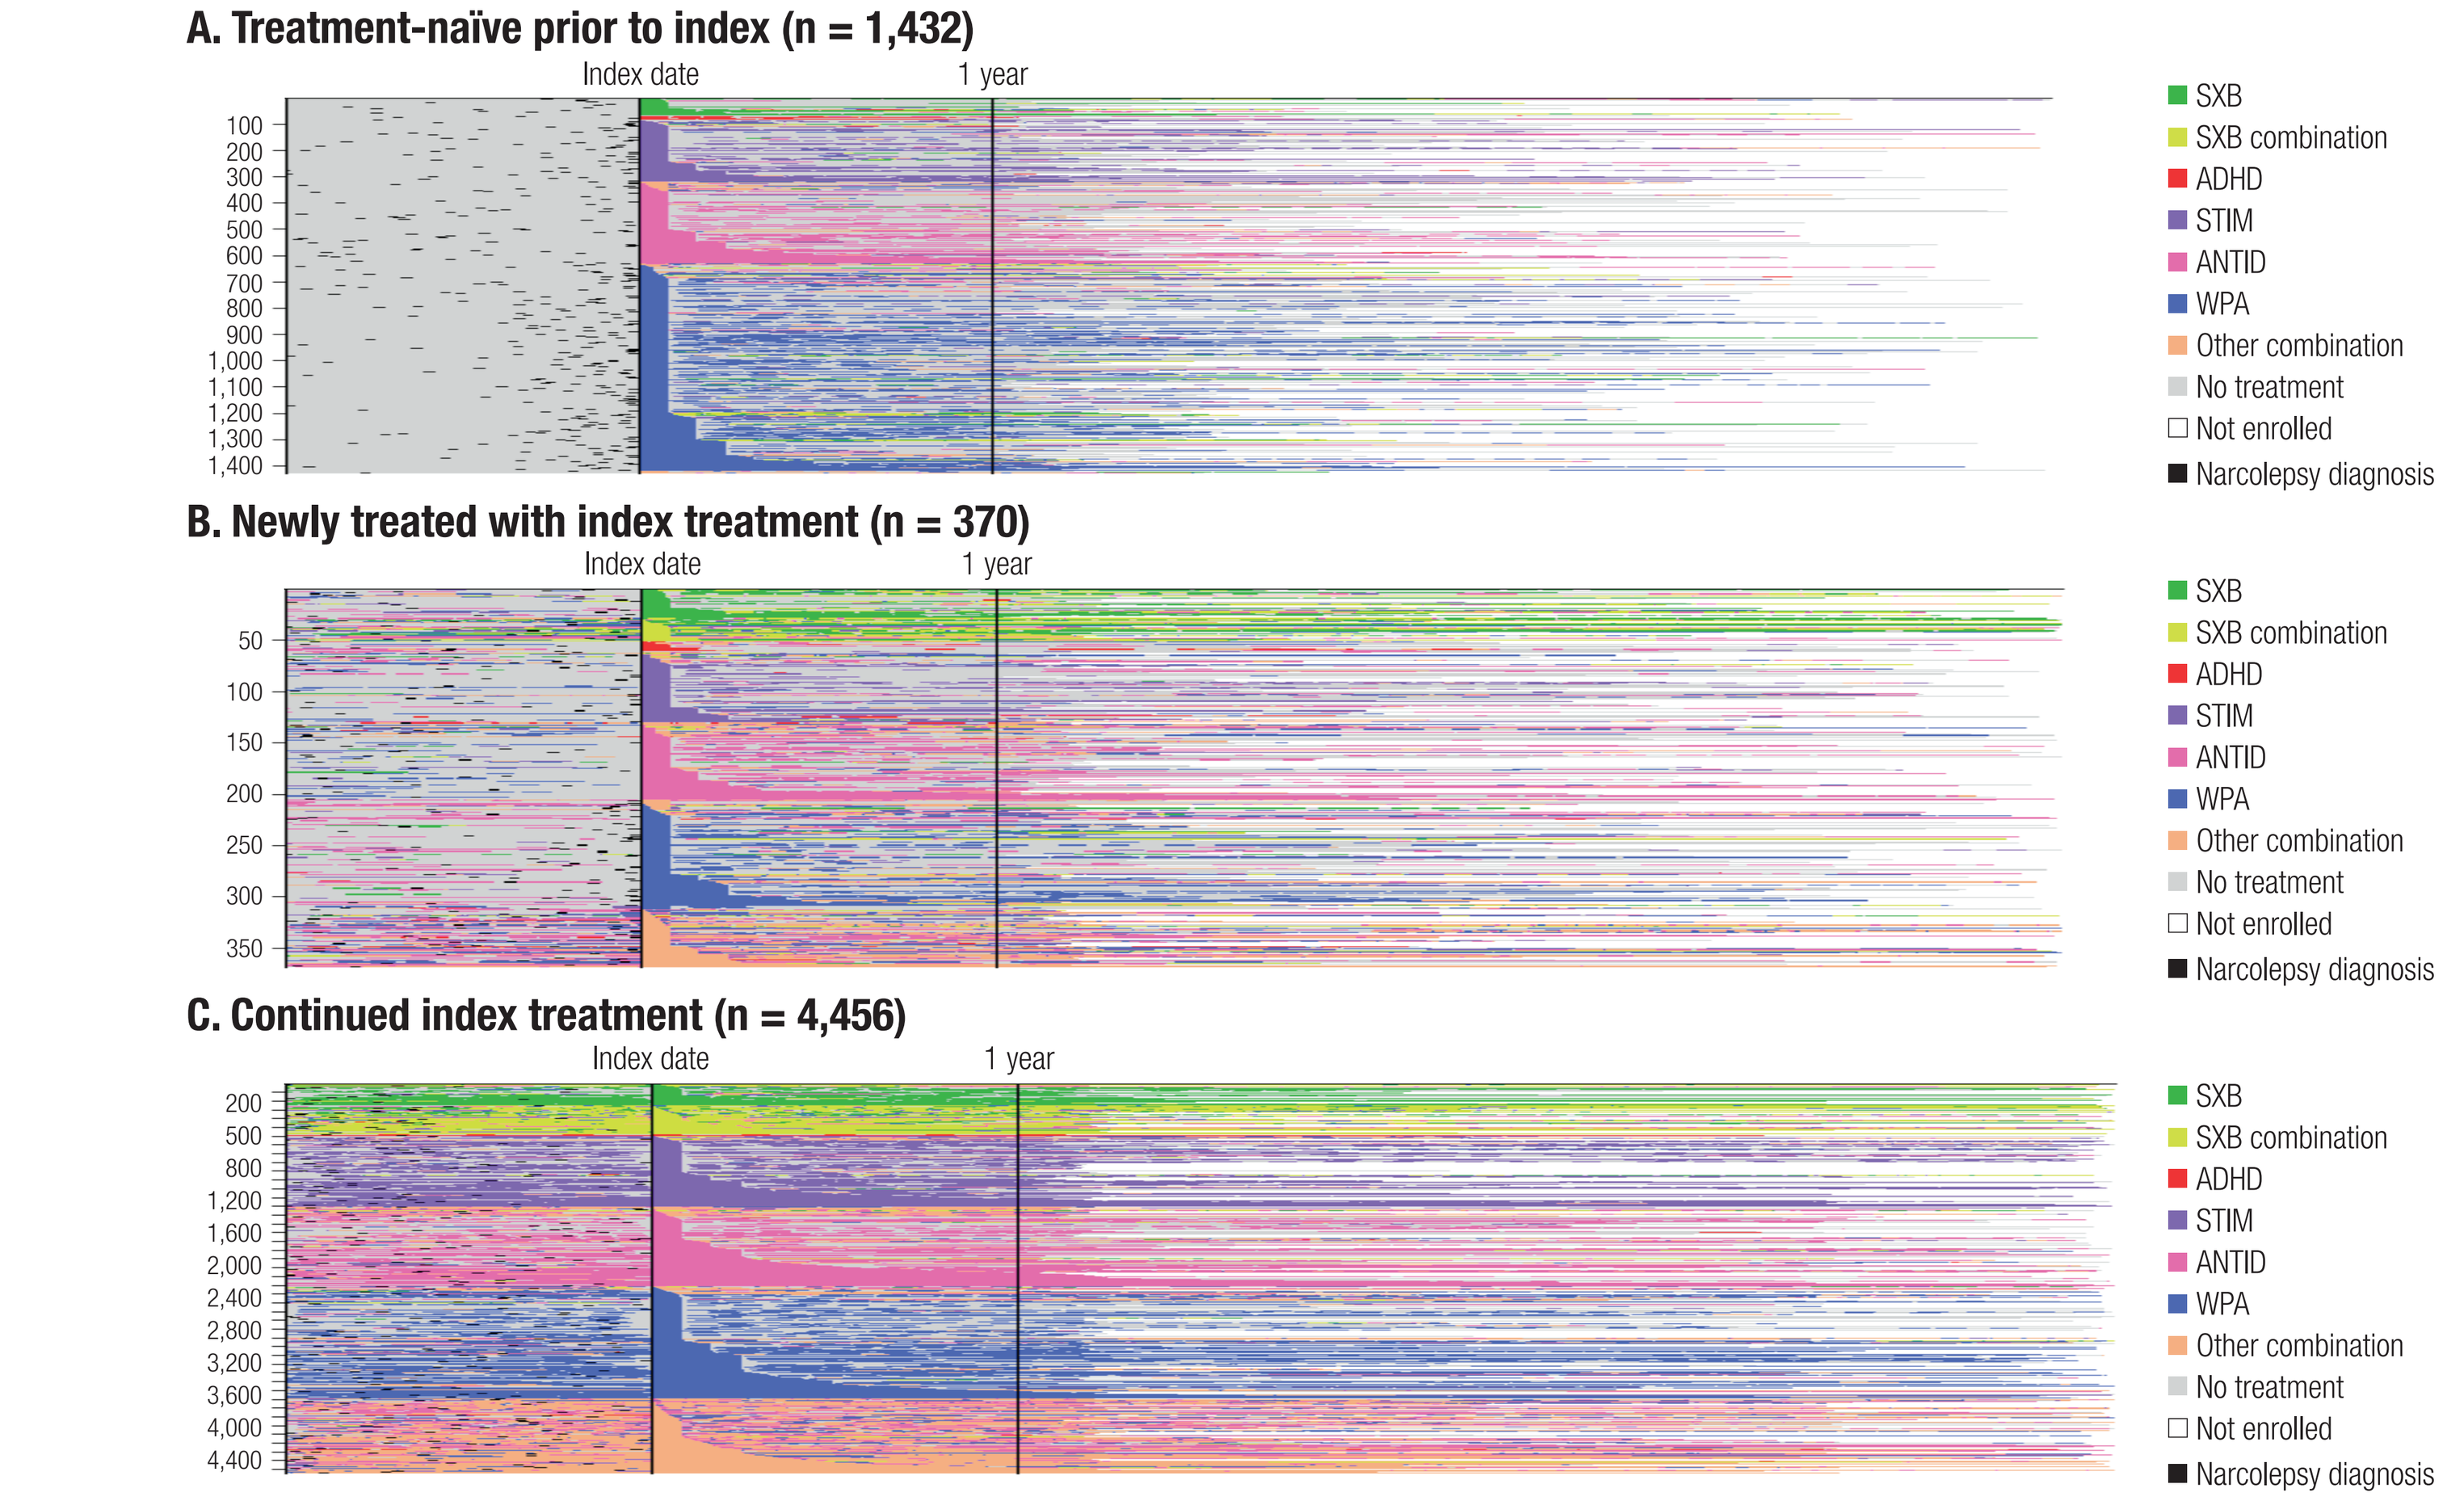
A. Treatment-naïve prior to index (n = 1,432)
B. Newly treated with index treatment (n = 370)
C. Continued index treatment (n = 4,456)
Index date
1 year
SXB
SXB combination
ADHD
STIM
ANTID
WPA
Other combination
No treatment
Not enrolled
Narcolepsy diagnosis

SXB, sodium oxybate; ADHD, attention-deficit/hyperactivity disorder; STIM, stimulant; ANTID, antidepressant; WPA, wake-promoting agent.
[a]Multicolored line segments represent different treatments received since the index date. The presence and length of segments and changes in colors and patterns over time indicate various treatment episodes. Each horizontal line represents an individual patient's treatment history in the follow-up period, and the height of each colored section is proportional to the total number of patients receiving each treatment. Gray areas represent periods of enrollment during which there were no claims for the treatments of interest.

- In all treatment groups (**Figures 5A-C**), interruptions in treatment use were common in the first year after the index date; however, many patients, including those taking SXB, appeared to resume the treatment at index date after an interruption, indicating persistence in use
 - In contrast, switching patterns seemed infrequent across treatments at the index date (**Figures 5A-C**)
- Adherence to the treatments at the index date (uninterrupted line segment) appeared more common among those who continued the treatment at the index date (**Figure 5C**) compared to treatment-naïve patients (**Figure 5A**) and those newly treated with index treatment (**Figure 5B**), which may suggest improved tolerability or dosing adjustment after continued exposure to treatment; this pattern was sharpest among users taking SXB
- For treatment-naïve patients (**Figure 5A**), the most common treatment at the index date was a WPA, followed by an antidepressant, an ADHD stimulant, and SXB

Limitations

- The study population primarily represents a commercially insured segment of the US population; results may not be generalizable to all patients with narcolepsy
- Claims-based analyses are limited by the possibility of diagnosis coding errors; however, concomitant requirements for MSLT and narcolepsy diagnosis codes may reduce this risk

Conclusions

- In real-world settings in the United States, SXB adherence and persistence rates were generally high in adult patients with narcolepsy over the 1-year assessment period
- High rates of adherence (mean PDC, 52%-86% and PDC ≥80%, 36%-74%) and persistence were observed for new and continuing SXB users relative to rates reported in the literature for other treatments[6-8]
 - Adherence for stimulants and nonstimulants for ADHD (1-year mean medication possession ratio, 0.57 [stimulants] and 0.49 [nonstimulants])[6]
 - Adherence for antidepressants for major depressive disorder (PDC ≥80% at 1 year, 26%)[8]
- Adherence and persistence rates were lower for new users than for continuing users
- These findings are consistent with data from clinical trials showing that titration to an optimal SXB dose, which may involve multiple steps, is critical in achieving an optimal response, which may take up to 2 months.[4,9-11] Moreover, adverse events with SXB are known to occur early and diminish over time.[11] Once optimal dosing is established, as with continuing users in this study, patients can achieve and maintain long-term treatment benefits[9,11-13]

References: 1. Thorpy MJ, Krieger AC. *Sleep Med.* 2014;15(5):502-507. **2.** Morgenthaler TI, et al. *Sleep.* 2007;30(12):1705-1711. **3.** Plazzi G, et al. *Lancet Child Adolesc Health.* 2018;2(7):483-494. **4.** Plazzi G, et al. *Sleep.* 2019;42(suppl 1):A307-A308. **5.** Mayer G, et al. *Sleep.* 2018;41(9). **6.** Gajria K, et al. *Neuropsychiatr Dis Treat.* 2014;10:1543-1569. **7.** Yeaw J, et al. *J Manag Care Pharm.* 2009;15(9):728-740. **8.** Keyloun KR, et al. *CNS Drugs.* 2017;31(5):421-432. **9.** Bogan RK, et al. *J Clin Sleep Med.* 2015;11(4):427-432. **10.** Thorpy MJ, et al. *Sleep Med.* 2015;16(1):9-18. **11.** XYREM® (sodium oxybate) oral solution [package insert]. Palo Alto, CA: Jazz Pharmaceuticals, Inc.; 2018. **12.** Mignot E, et al. *Sleep.* 2018;41(suppl 1):A302. **13.** Mignot E, et al. *Sleep.* 2019;42(suppl 1):A308.

Support: This study was sponsored by Jazz Pharmaceuticals.
Acknowledgements: Writing and editorial support were provided by Lauren Fink, PhD, of MedErgy, and were funded by Jazz Pharmaceuticals.
Disclosures: AMH is a consultant for UCB Pharmaceuticals, Jazz Pharmaceuticals, Biogen Idec, Sage Therapeutics, Eisai Pharmaceuticals, Marinus Pharmaceuticals, and Neurelis Pharmaceuticals; has received research support from UCB Pharmaceuticals, Biogen Idec, Sage Therapeutics, and Marinus Pharmaceuticals; has received royalties from Springer Publishing Company, Demos Medical Publishing, and Wolters Kluwer; and has an editorship with Wolters Kluwer. **CMR** has no disclosures to report. **SRR** and **RST** are employees of Partnership for Health Analytic Research, LLC, which received funding from Jazz Pharmaceuticals to conduct this study. **DH**, **JP**, and **MY** are full-time employees of Jazz Pharmaceuticals, who, in the course of this employment, have received stock options exercisable for, and other stock awards of, ordinary shares of Jazz Pharmaceuticals, plc. **MB** and **KFV** are former employees of Jazz Pharmaceuticals, who, in the course of this employment, received stock options exercisable for, and other stock awards of, ordinary shares of Jazz Pharmaceuticals, plc.

Scan this code to access this poster online
This code is not for promotional purposes.