A4094

Rates of Adherence and Persistence of Antifibrotic Therapies in the US Medicare Population

Mitra Corral,[1] Sheila R. Reddy,[2] Eunice Chang,[2] Michael S. Broder,[2] Sohum Gokhale,[2] Karina Raimundo[1]

[1]Genentech, Inc., South San Francisco, CA; [2]Partnership for Health Analytics, LLC, Beverly Hills, CA

BACKGROUND

- Idiopathic pulmonary fibrosis (IPF) is a progressive, irreversible, fibrotic lung disease with a poor prognosis[1-2]
- Pirfenidone and nintedanib are 2 antifibrotic therapies approved by the Food and Drug Administration (FDA) in October 2014 for the treatment of IPF[3-4]
 - Both therapies slow disease progression, as measured by changes in lung function defined by forced vital capacity[5-7]
- Adherence to and persistence with antifibrotic therapy are important because they enable patients to observe one of the primary benefits of treatment: delaying IPF progression
- Previous studies with Medicare data have described the incidence and prevalence of IPF, as well as its disease course[8]
- Limited real-world data are available about adherence to and persistence with antifibrotic therapies

OBJECTIVE

- To examine adherence to and persistence with antifibrotic therapies in a US Medicare population

METHODS

Patients

- A retrospective observational study of Medicare beneficiaries using the 100% Medicare Research Identifiable File was conducted from October 2014 through December 2015
- Patients were included if they had the following:
 - Age ≥ 67 years on index date (initiation of antifibrotic therapy) to allow for 2 years of observation prior to index (baseline period) and the identification of patients with newly diagnosed disease ***and***
 - ≥ 1 inpatient or outpatient claim with IPF listed as a diagnosis (ICD-9-CM: 516.3, 516.30 and 516.31; ICD-10-CM: J84.111, J84.112) during the study period, occurring on or before the index date ***and***
 - ≥ 1 fill of pirfenidone or nintedanib during the identification period (October 15, 2014 [FDA approval date] to December 31, 2015), the first date of which was the index date, and no use in the 2 years prior to the index date
- Key exclusion criteria were a lung transplant prior to index and no eligibility for or continuous enrollment in the fee-for-service Medicare Parts A and B or Medicare Part D (and those enrolled in Medicare Advantage) in the 2 years before index
- The cohort was divided into 2 groups: initiators of pirfenidone and initiators of nintedanib

Outcomes

- Adherence was defined as a proportion of days covered (PDC) ≥ 0.80
 - PDC was calculated by dividing the number of days in which medication was available to the patient by the total number of days of follow-up
- Persistence was defined as the duration in use from treatment initiation to discontinuation (i.e., stop in therapy after a gap in use ≥ 60 days or switch to other antifibrotic therapy), lung transplant, disenrollment or death. Persistence was reported as follows:
 - Proportion of patients who used index therapy continuously (without gap in use of ≥ 60 days or switch) until end of follow-up
 - Proportion of patients who stopped index therapy by the end of follow-up
 - Proportion of patients who switched from index therapy by the end of follow-up
 - Time to discontinuation

Statistical Analyses

- Demographics, clinical characteristics and adherence to antifibrotic therapy were characterized using descriptive statistics
 - T-tests were used for continuous variables (means) and χ^2 tests were used for categorical variables (percentages)
- Time to discontinuation was assessed by Kaplan-Meier analysis and compared between groups using the log-rank test

RESULTS

Study Population

- In total, 3546 patients who received a diagnosis of IPF initiated antifibrotic therapy and met the study criteria (**Table 1**)
 - 2082 patients received pirfenidone
 - 1464 patients received nintedanib
- The majority of patients in both groups were male and white, with mean age > 75 years

Table 1. Baseline Patient Demographics and Clinical Characteristics of Patients With IPF Receiving Antifibrotics in a US Medicare Population

Characteristic	Pirfenidone (n = 2082)	Nintedanib (n = 1464)	*P* Value
Age, mean (SD), years	75.6 (5.5)	76.3 (5.9)	< 0.001
Female, n (%)	694 (33.3)	593 (40.5)	< 0.001
White, n (%)	1985 (95.3)	1397 (95.4)	0.908
Region, n (%)			< 0.001
Midwest	507 (24.4)	389 (26.6)	
Northeast	384 (18.4)	207 (14.1)	
South	798 (38.3)	670 (45.8)	
West	393 (18.9)	198 (13.5)	
Newly diagnosed IPF, n (%)*	1059 (50.9)	842 (57.5)	< 0.001
Oxygen therapy in previous year, n (%)	1743 (83.7)	1215 (83.0)	0.189
Pulmonary rehabilitation in previous year, n (%)	1985 (95.3)	1388 (94.8)	0.469
CT scan in previous year, n (%)	1536 (73.8)	1133 (77.4)	0.014
Distance from patient residential area to an ILD center, mean (SD), miles	100 (175)	98 (138)	0.661

CT, computed tomography; ILD, interstitial lung disease; IPF, idiopathic pulmonary fibrosis.
* ≥ 1 year disease-free in pre-index period.

- Comorbidities were common between the pirfenidone and nintedanib groups, including chronic obstructive pulmonary disease, gastroesophageal reflux disease and cardiovascular conditions (**Table 2**)

Table 2. Baseline Comorbidities in Patients With IPF Receiving Antifibrotics in a US Medicare Population

Characteristic	Pirfenidone (n = 2082)	Nintedanib (n = 1464)	*P* Value
Charlson Comorbidity Index, mean (SD)	3.9 (3.0)	4.2 (3.0)	0.014
Charlson Comorbidity Index excluding COPD, mean (SD)	3.2 (3.9)	3.4 (2.9)	0.031
Number of chronic conditions, mean (SD)	7.7 (2.0)	7.8 (2.0)	0.031
Condition, n (%)			
Cor pulmonale	106 (5.1)	83 (5.7)	0.450
COPD, including emphysema	1187 (57.0)	895 (61.1)	0.014
Pneumonia (bacterial or viral)	700 (33.6)	566 (38.7)	0.002
GERD	1230 (59.1)	863 (58.9)	0.938
Obstructive sleep apnea	627 (30.1)	431 (29.4)	0.665
Obesity	501 (24.1)	348 (23.8)	0.841
Lung cancer	56 (2.7)	50 (3.4)	0.212
Pneumothorax	163 (7.8)	91 (6.2)	0.067
Smoking cessation therapy	73 (3.5)	54 (3.7)	0.774
Any cardiovascular condition, n (%)	1508 (72.4)	1066 (72.8)	0.801
Pulmonary hypertension	254 (12.2)	178 (12.2)	0.970
Ischemic heart disease	1180 (56.7)	839 (57.3)	0.708
Congestive heart failure	608 (29.2)	448 (30.6)	0.370
Venous thromboembolism	203 (9.8)	143 (9.8)	0.986
Stroke	147 (7.1)	130 (8.9)	0.047
Atrial fibrillation	500 (24.0)	319 (21.8)	0.122

COPD, chronic obstructive pulmonary disease; GERD, gastroesophageal reflux disease.

- Patients treated with pirfenidone had a longer duration of follow-up than patients treated with nintedanib
 - The mean (SD) duration of follow-up for the pirfenidone and nintedanib groups was 204.1 (97.7) days and 188.6 (113.6) days, respectively (t-test, $P < 0.001$)

Adherence to and Persistence With Antifibrotic Therapy

- Adherence was significantly higher in patients receiving pirfenidone vs. nintedanib (mean PDC, 0.79 vs. 0.75, respectively; $P < 0.001$; **Table 3**)
- Persistence, indicated by percentage still receiving index treatment at the end of follow-up, was significantly higher in patients treated with pirfenidone vs. nintedanib (75.4% vs. 70.9%; $P < 0.001$)
- A higher proportion of patients receiving pirfenidone vs. nintedanib switched therapy (5.1% vs. 3.1%, respectively)

Table 3. Adherence to and Persistence With Antifibrotic Therapies in Patients With IPF in a US Medicare Population

	Pirfenidone (n = 2082)	Nintedanib (n = 1464)	*P* Value
Index antifibrotic ending status, n (%)			< 0.001
Continuation*	1570 (75.4)	1038 (70.9)	
Discontinuation			
Stopping†	406 (19.5)	381 (26.0)	
Switching‡	106 (5.1)	45 (3.1)	
PDC during follow-up, mean (SD)§	0.79 (0.26)	0.75 (0.30)	< 0.001
PDC ≥ 0.80, n (%)	1370 (65.8)	886 (60.5)	0.001

PDC, proportion of days covered.
* Still receiving index treatment at the end of follow-up.
† With a gap ≥ 60 days.
‡ To a non-index antifibrotic, without a ≥ 60-day gap of index treatment.
§ The follow-up time varied among patients.

- At Day 365, the probability of continuing antifibrotic therapy was statistically significantly greater in patients receiving pirfenidone vs. nintedanib (**Figure 1**)
- Similarly, discontinuation rates over follow-up were lower for patients receiving pirfenidone vs. nintedanib (19.5% vs. 26.0%, respectively) (**Table 3**)

Figure 1. Time to Discontinuation in Patients Receiving Antifibrotic Therapy in a US Medicare Population

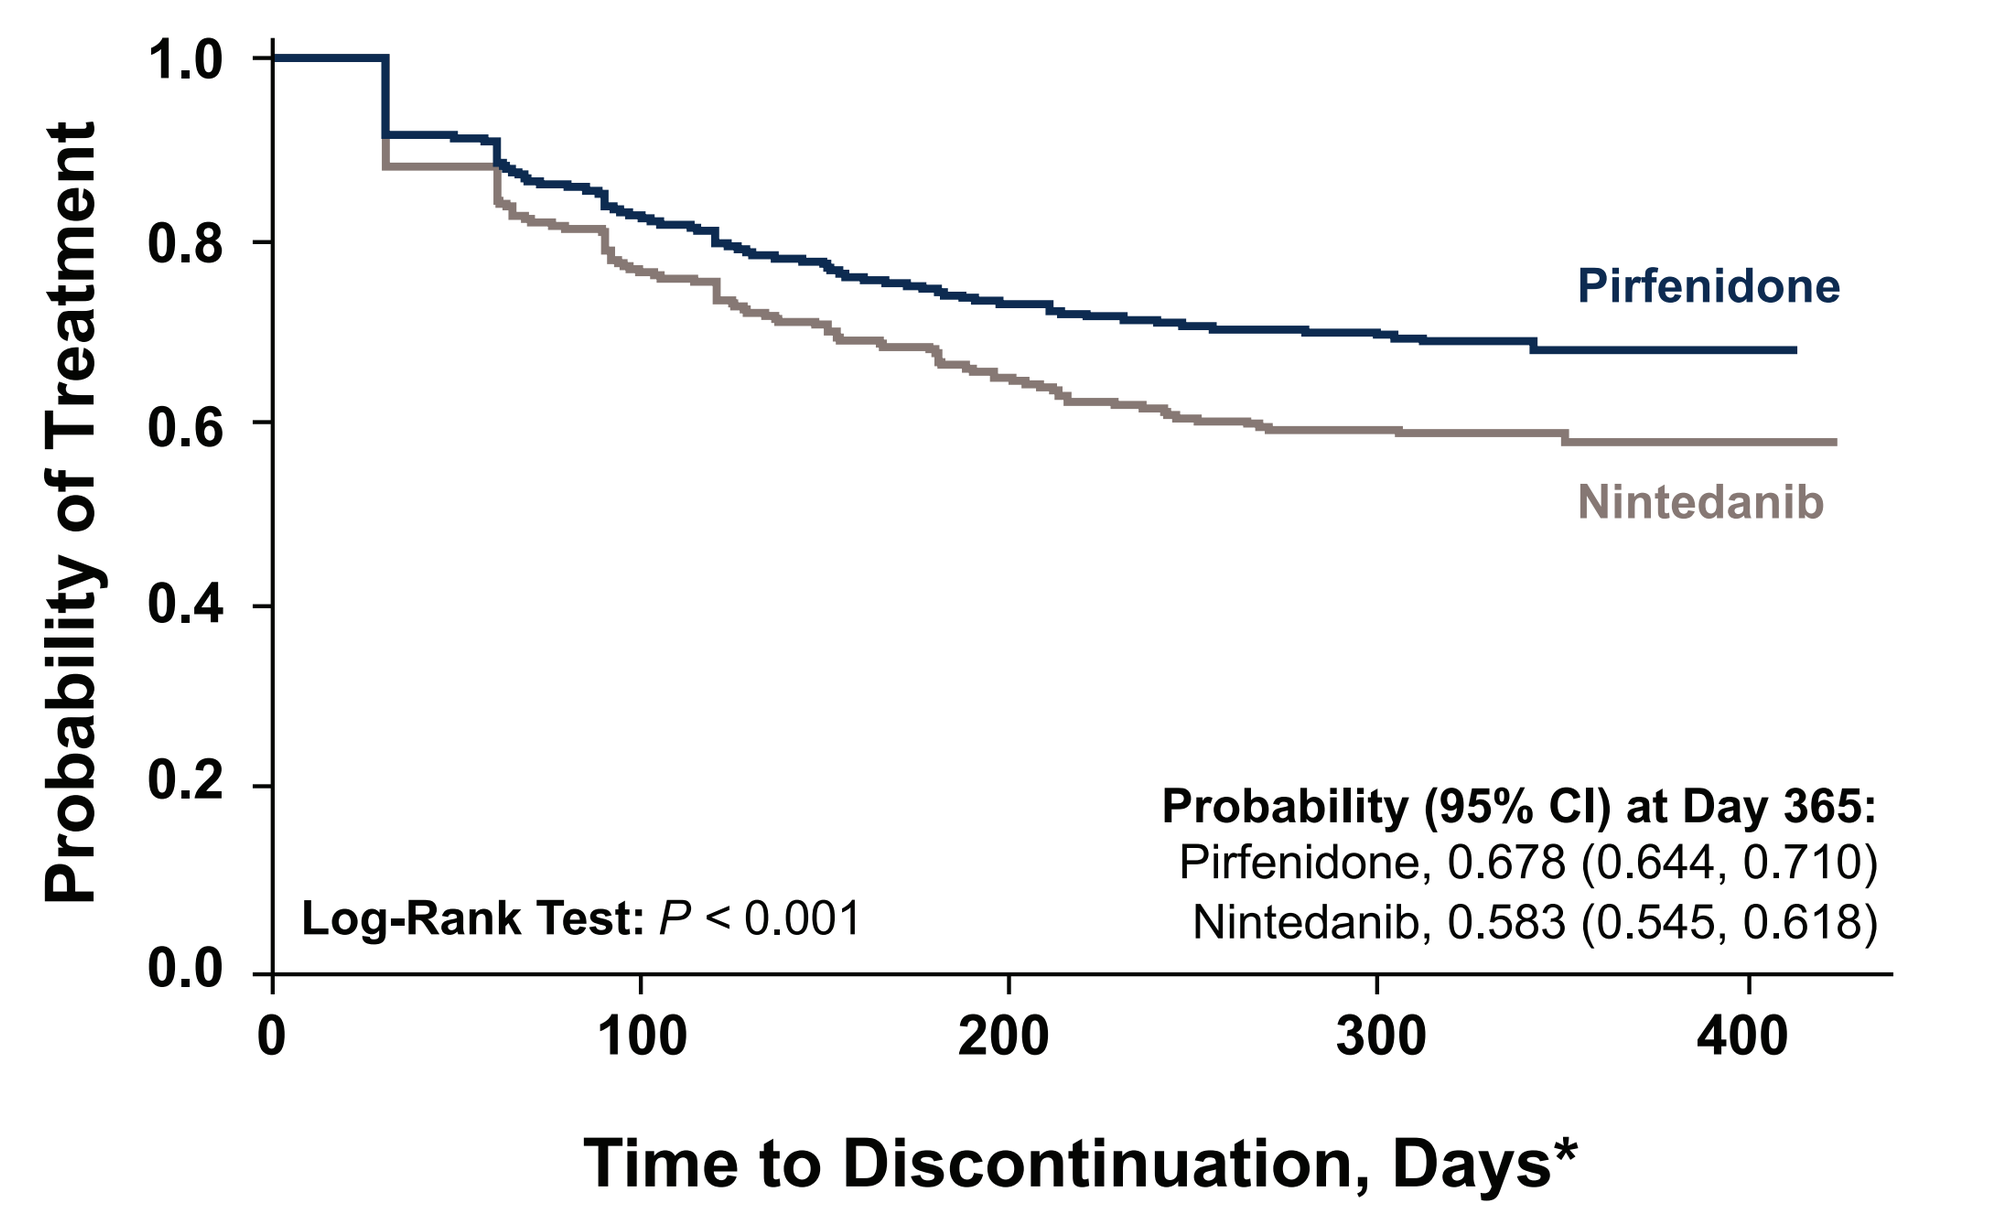
Probability of Treatment
1.0
0.8
0.6
0.4
0.2
0.0
Pirfenidone
Nintedanib
Probability (95% CI) at Day 365:
Pirfenidone, 0.678 (0.644, 0.710)
Nintedanib, 0.583 (0.545, 0.618)
Log-Rank Test: P < 0.001
0
100
200
300
400
Time to Discontinuation, Days*

Number at risk:	Day 0	Day 90	Day 180	Day 270	Day 365	Day 428
Pirfenidone	2082	1450	996	530	28	0
Nintedanib	1464	870	464	230	56	0

* Discontinuation defined as stopping index antifibrotic with a gap ≥ 60 days or switching to a non-index antifibrotic. Patients were censored at the time of transplant, death, study end or disenrollment, whichever occurred first.

LIMITATIONS

- These results are unadjusted and do not account for potential differences between treatment groups; however, using a sensitivity analysis with inverse probability of treatment weighting to balance observed characteristics between the groups, nearly identical results were obtained
- As with other claims analyses, these findings on adherence and persistence reflect patterns in prescription fills (and not use) of the index medications; therefore, they may not fully capture actual utilization patterns of antifibrotics among patients with IPF

CONCLUSIONS AND IMPLICATIONS

- Adherence and persistence were both statistically significantly higher in patients with IPF who received pirfenidone vs. nintedanib during follow-up
- Fewer patients receiving pirfenidone stopped therapy and a higher percentage of patients receiving pirfenidone switched treatments during the follow-up period
- Further analysis may be needed to determine whether these trends continue over longer time periods

REFERENCES

1. Raghu G, et al. *Am J Respir Crit Care Med.* 2015;192:e3–e19.
2. Ley B, et al. *Am J Respir Crit Care Med*. 2011;183:431–440.
3. Esbriet (pirfenidone) [package insert]. South San Francisco, CA: Genentech, Inc.; 2017.
4. Ofev (nintedanib) [package insert]. Ridgefield, CT: Boehringer Ingelheim Pharmaceuticals; 2018.
5. Noble PW, et al. *Lancet*. 2011;377:1760–1769.
6. King TE Jr, et al. *N Engl J Med*. 2014;370:2083–2092.
7. Richeldi L, et al. *N Engl J Med*. 2014;370:2071–2082.
8. Raghu G, et al. *Lancet Respir Med*. 2014;2:566–572.

DISCLOSURES

Data within the poster are directly derived from research sponsored by Genentech, Inc.

M. Corral and K. Raimundo are employees of Genentech, Inc. S. R. Reddy, E. Chang, M. S. Broder and S. Gokhale are employees of Partnership for Health Analytic Research, LLC, a health services research company paid by Genentech, Inc. to conduct this research.

Presented at the American Thoracic Society 2019 International Conference; May 17–22, 2019; Dallas, TX.

This poster was sponsored by Genentech, Inc. and F. Hoffmann-La Roche Ltd.
Third-party writing assistance was furnished by Health Interactions, Inc.