How Accurate are Budget Impact Predictions for New Drugs? An Empirical Analysis

Michael S. Broder, MD, MSHS[1]; Jenelle M. Zambrano, DNP, CNS, RN[1]; Jackie Lee, BS[1]; Richard S. Marken, PHD[2]

[1] *Partnership for Health Analytic Research, LLC, Beverly Hills, CA;* [2] *RSM Consulting, Los Angeles, CA*

BACKGROUND

- Concern about the prices of newly launched drugs is rising among the public and among healthcare payers.
- Various media reports often quote estimates of the impact new drugs are expected to have on healthcare costs.
- The accuracy of these estimates has not been rigorously studied.

OBJECTIVE

- To evaluate the accuracy of pre-launch predictions of the budget impact of newly approved drugs.

METHODS

Study Design

We identified a sample of newly developed drugs that received FDA approval in the last 5 years. We searched for pre-launch budget impact estimates and post-launch actual budget impact for these drugs and compared them to evaluate accuracy. We grouped pre-launch estimates into those by individual analysts, which we termed “informal” analyses, and those that presented a summary of multiple analysts’ estimates, or were from governmental bodies, payers, or other large healthcare organizations, which we termed “formal.” We then examined factors that might affect the accuracy of the pre-launch budget impact estimates.

Study Measures

- Pre-launch budget impact estimate
 - *Sources:* Medical news publications, non-profit research organizations, and news reports
 - *Time frame:* Launch year-1 (e.g., if drug launch was in 2013, then 2012 was used)
 - *Keywords:* Trade/generic name, estimated sales, predicted sales, revenue, annual sales, sales forecast, launch year+1, report, US, United States
- Post-launch actual budget impact (sales)
 - *Source:* Sales revenue from manufacturers’ earnings reports
 - *Time frame:* year of predicted sales estimate (e.g., if pre-launch estimate predicted sales for 2015, then 2015 was used)
- Ratio of Predicted Cost (P) to Actual Cost (A): P/A
 - *Predicted cost* = pre-launch budget impact estimate of a drug
 - *Actual cost* = post-launch actual budget impact of a drug

Statistical Analysis

- P/A determined the degree of over or under estimation of predicted to actual budget impact. Predicted cost was an accurate estimate of actual Cost if P/A = 1.

RESULTS

- We identified 24 pre-launch estimates for 14 drugs.
- Multiple budget impact estimates were found for several drugs (Opdivo, Viekira Pak, Repatha, Praluent, PCSK9 combined, Cosentyx, and Solvaldi).
- Predicted and actual costs are shown in **Fig 1**.

Figure 1: Pre-launch budget impact estimate versus actual budget impact by drug and year of pre-launch estimate*

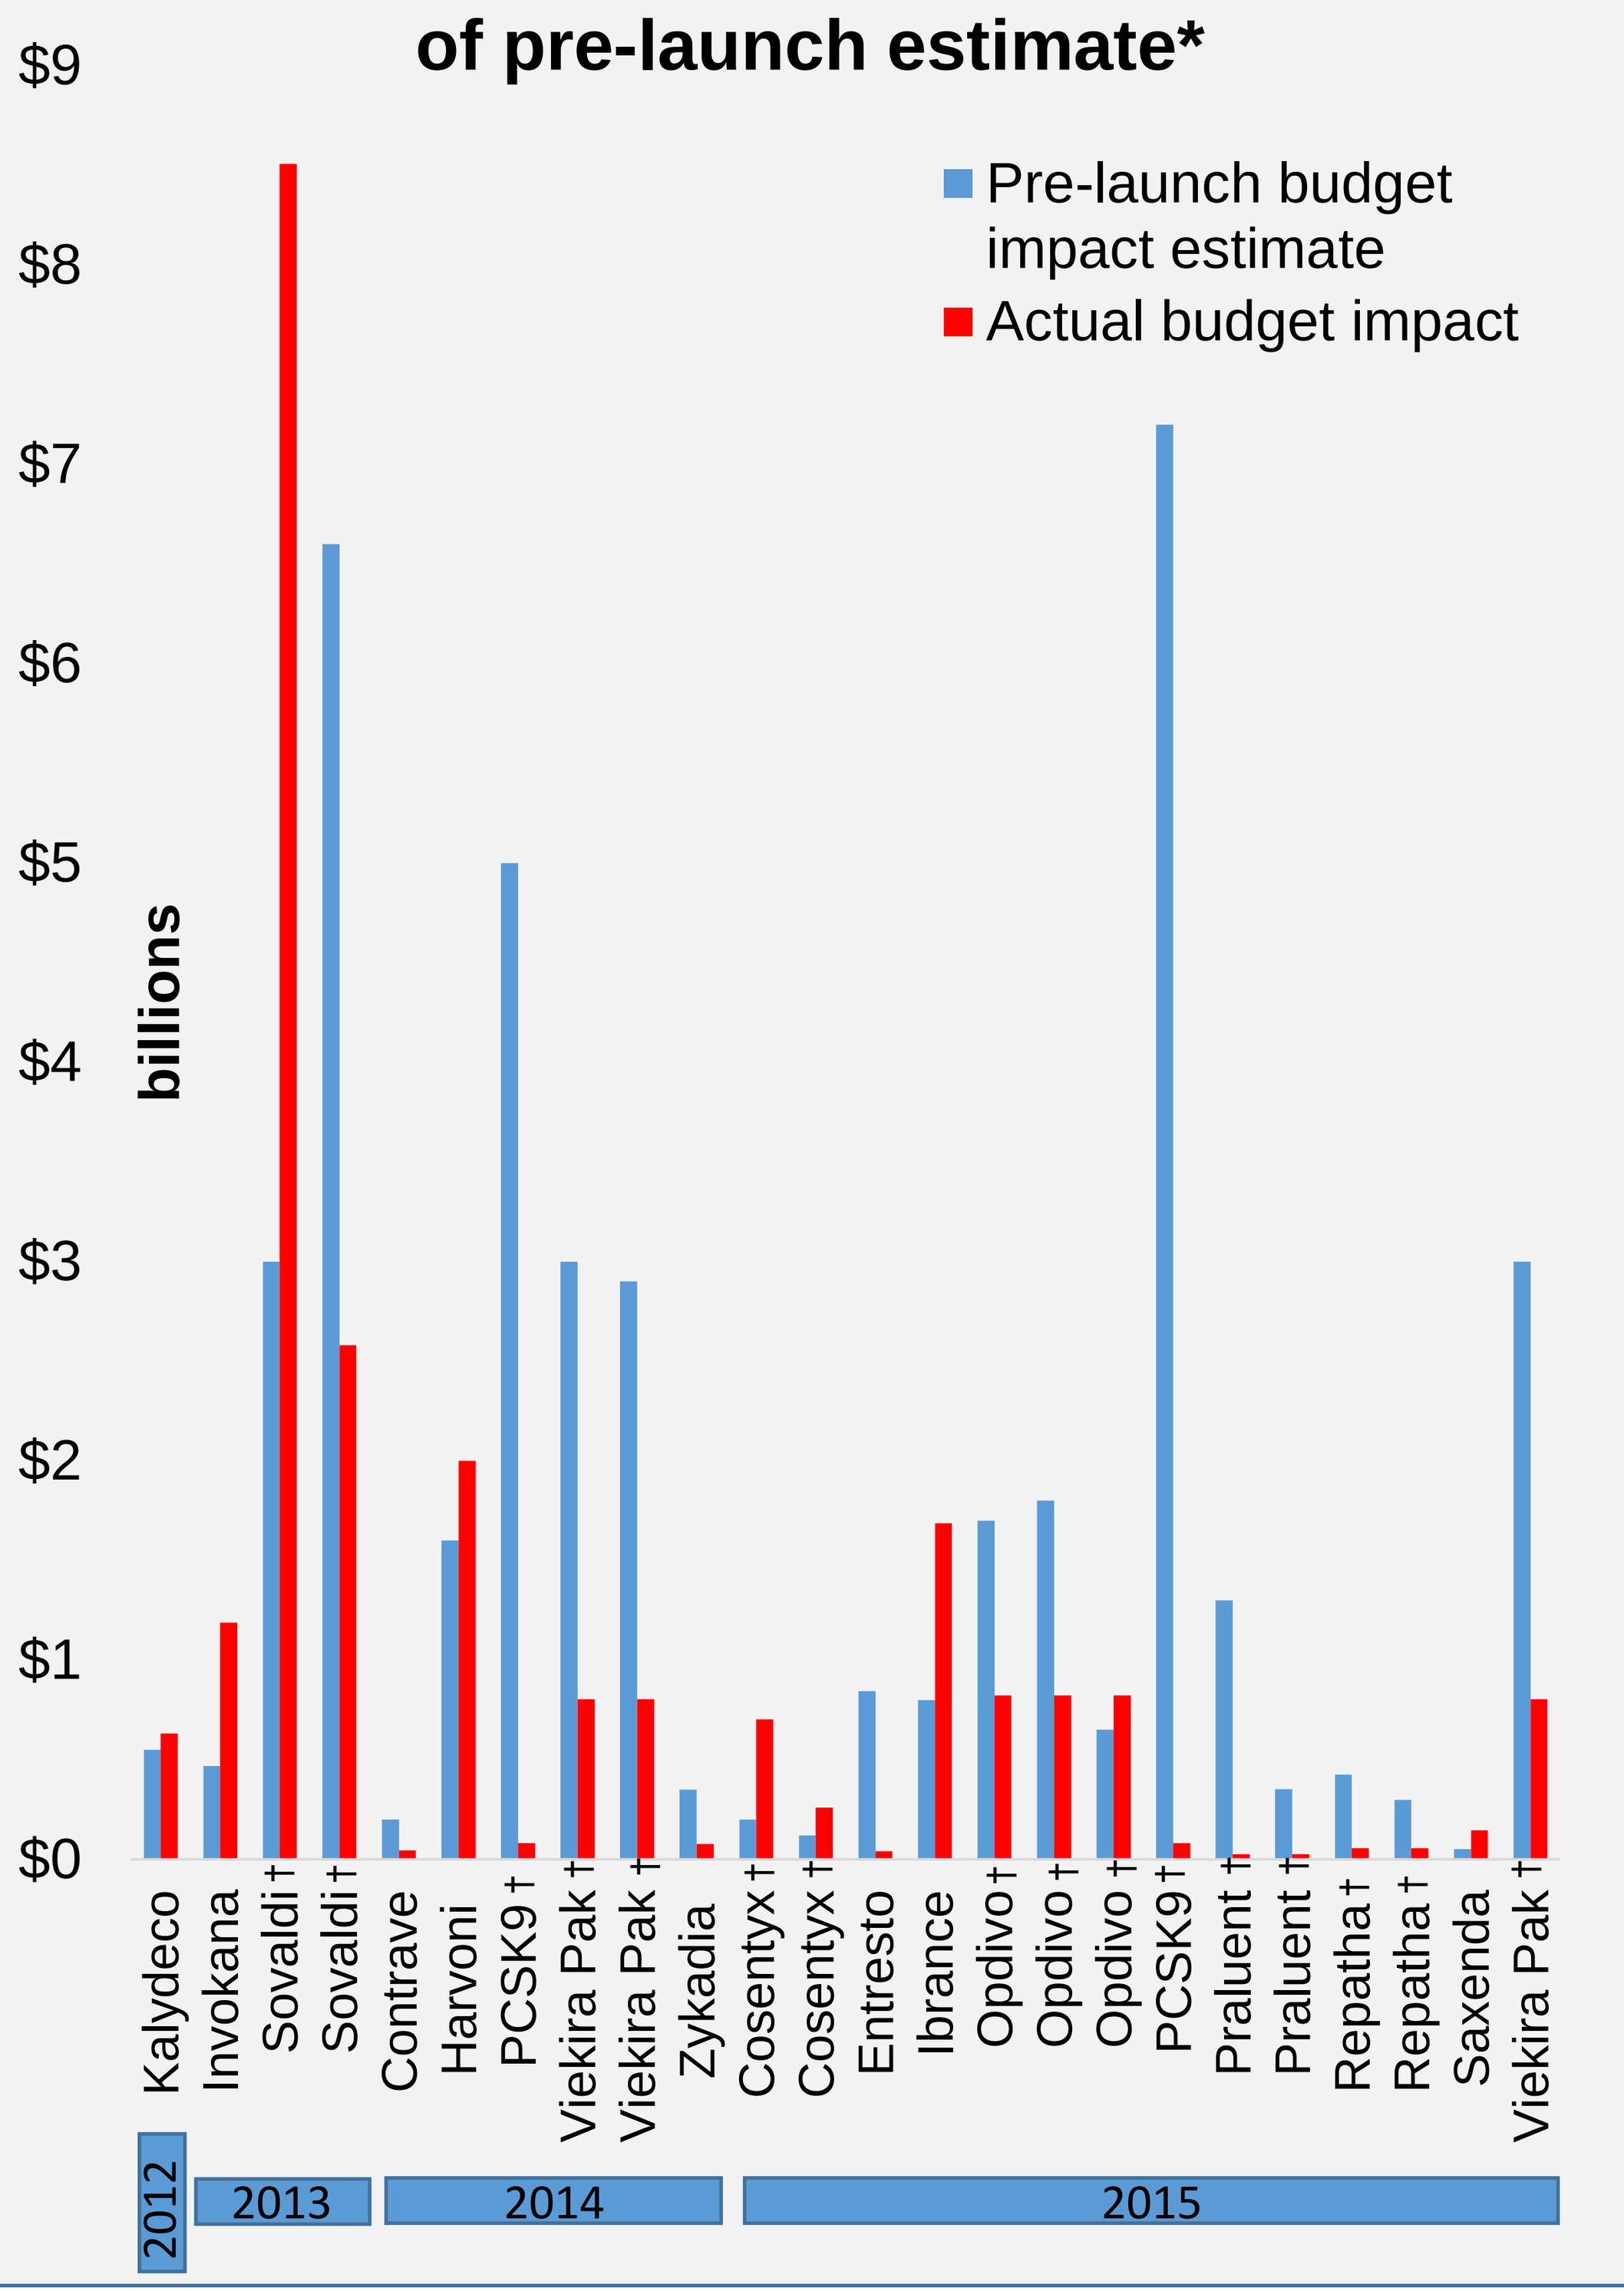
$9
$8
$7
$6
$5
$4
$3
$2
$1
$0
billions
Pre-launch budget impact estimate
Actual budget impact
Kalydeco
Invokana
Sovaldi †
Sovaldi †
Contrave
Harvoni
PCSK9 †
Viekira Pak †
Viekira Pak †
Zykadia
Cosentyx †
Cosentyx †
Entresto
Ibrance
Opdivo †
Opdivo †
Opdivo †
PCSK9 †
Praluent †
Praluent †
Repatha †
Repatha †
Saxenda
Viekira Pak †
2012
2013
2014
2015

* Pre-launch estimate year indicates the year sales estimate was made.
† Multiple estimates for the same drug or group of drugs.

RESULTS (CONT.)

- Mean prediction was 11.4 times higher than actual cost, median 3.1 times higher **(Table 1)**.

Table 1: Accuracy, Measured by Predicted Cost/Actual Cost

	N	Mean	(SD)	[Median]
Overall	24	11.4	(22.0)	[3.1]
Formal[1]	16	12.3	(23.2)	[3.1]
Informal[2]	8	9.4	(20.6)	[2.3]

[1] Methods described or using aggregated results of multiple unrelated estimates.
[2] Estimates reported by individual analysts.

- Predicted cost varied between 0.3 and 87 times actual cost **(Fig 2)**.

Figure 2: Distribution of predicted and actual budget impact ratios of 14 evaluated drugs

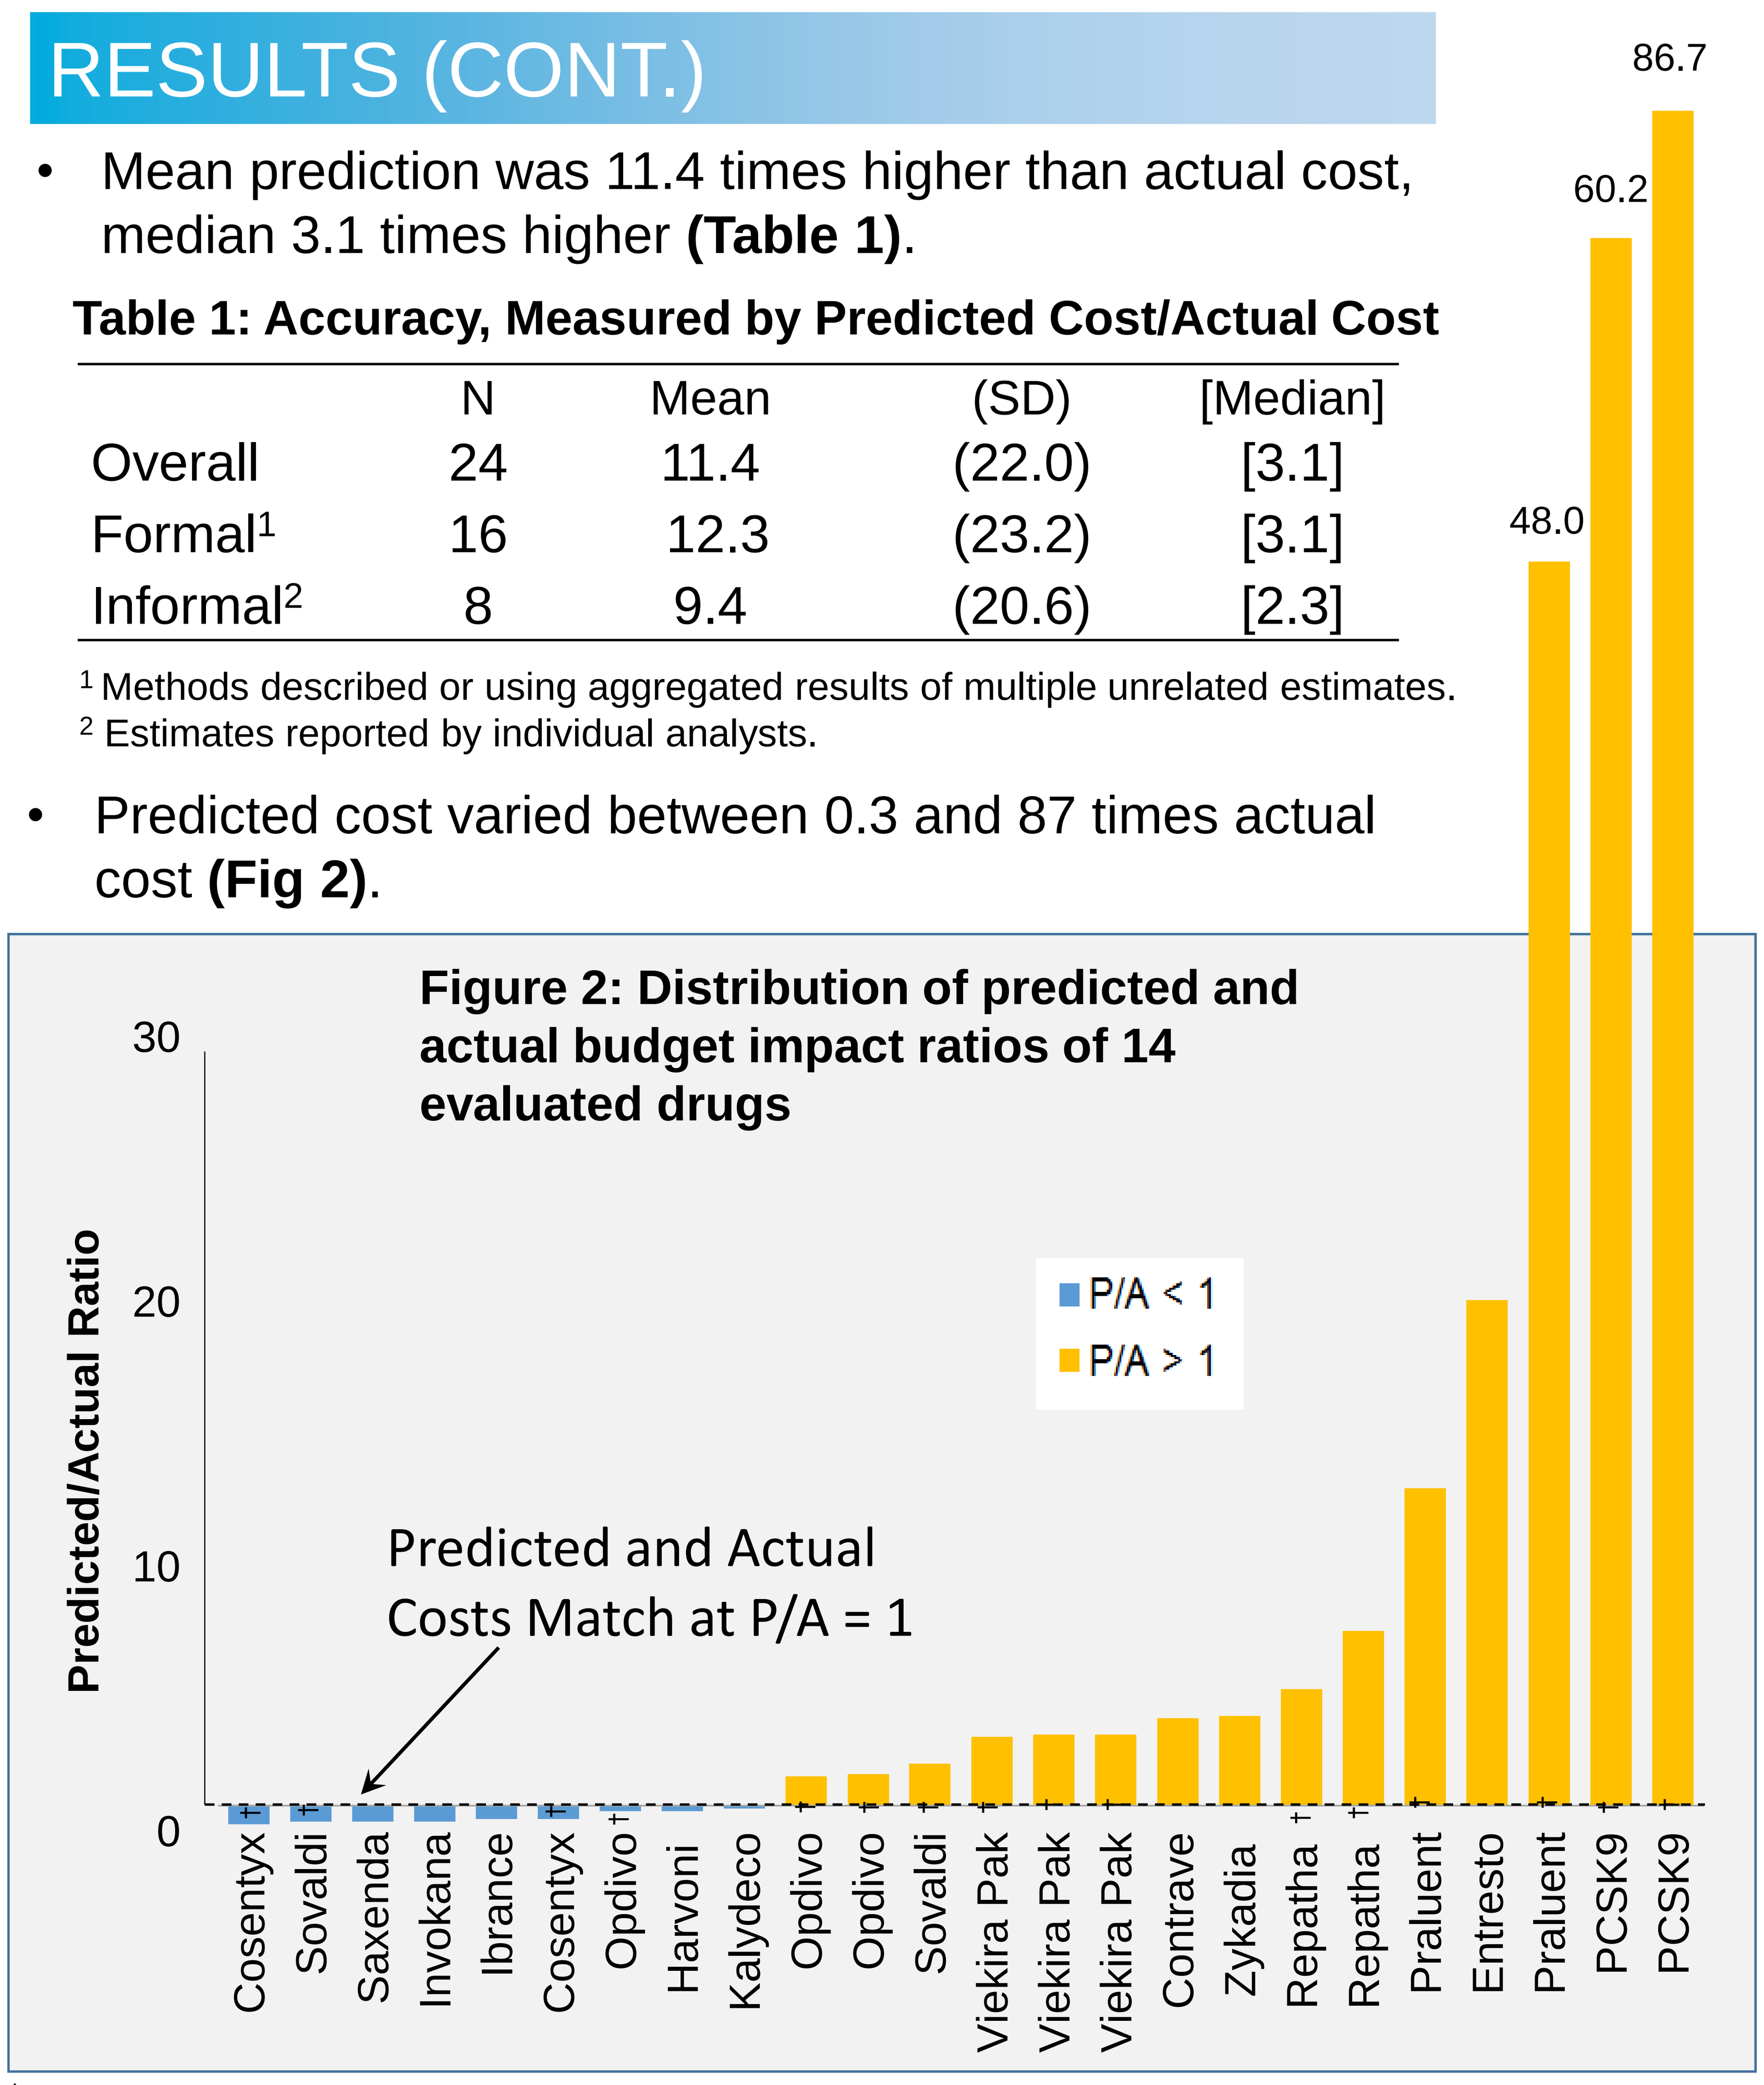
86.7
60.2
48.0
30
20
10
0
Predicted/Actual Ratio
P/A < 1
P/A > 1
Predicted and Actual
Costs Match at P/A = 1
Cosentyx †
Sovaldi †
Saxenda
Invokana
Ibrance
Cosentyx †
Opdivo †
Harvoni
Kalydeco
Opdivo †
Opdivo †
Sovaldi †
Viekira Pak †
Viekira Pak †
Viekira Pak †
Contrave
Zykadia
Repatha †
Repatha †
Praluent †
Entresto
Praluent †
PCSK9 †
PCSK9 †

† Multiple estimates for the same drug or group of drugs.

- P/A budget impact ratios were most inaccurate for PCSK9-combined and Praluent.
 - The Institute for Clinical and Economic Review estimated $7.2 billion for PCSK9's first year of sales. Actual sales were $0.08 billion (based on initial 2 quarters), around 1% of predicted.
 - PwC Health forecasted $1.3 billion for Praluent's US sales; actual sales were $0.03 million or 2.1% of predicted.
- One source of error was that the proportion of patients who actually used the drug was much lower than what was assumed; another was the failure to account for the discounting of drug prices.
- The largest underestimate was for Sovaldi, with $3 billion predicted and $8.5 billion in actual sales.

CONCLUSIONS

- For a sample of recently launched drugs, the pre-launch predictions of their budget impact were considerable overestimates of their actual budget impact, although there were some notable underestimates.
- For every $11 of predicted cost, there was $1 of actual cost to the healthcare system.
- Neither drug price nor source of budget impact estimate appeared to be associated with the accuracy of the estimate.
- Using an older but larger data set and slightly different methodology, a prior study focusing only on equity analysts’ forecasts found forecasts were commonly more than 200% of actual sales.[1]
- Potential reasons for this systematic bias may include over-optimism on the part of analysts.[2,3]
- Bias may also result from the desire to maximize the media impact of pre-launch sales estimates.
- Overestimating budget impact may lead to early access restrictions, higher copays, and other changes in plan design that would ultimately impact patients.
- Predictions of the cost impact of new drugs should be constantly evaluated in order to improve them.

REFERENCES

1. Cha, M., et al. (2013) Pharmaceutical forecasting: Throwing darts? *Nature Reviews*, 12:737-8.
2. Capps, G. (2016) Natural optimism in financial reporting. *Accounting Horizons*, 30(1):79-91.
3. Lin, W-C., et al. (2013) The over-optimism of financial analysts and the long-run performance of firms following private placements of equity. *Finance Research Letters,* 10(2):82–92.

Presented at AMCP NEXUS, October 3-6, 2016, National Harbor, MD

Research was conducted by Partnership for Health Analytic Research, LLC.
This study was sponsored by Amgen.